Modulation of IL-12p70 exposure and activity following sequential administration of tumor targeted self-assembling split IL-12 subunits

Joshua Heiber[1], Pinar Gurel[1], Robert G. Newman[1], Kristiana Dreaden[1], Yanchun Zhao[1], Chunhua Wang[1], Su-Ping Pearson[1], Jean Chamoun[1]

[1]Mural Oncology, Waltham, MA, USA

Take Away

- Mural Oncology has developed an innovative approach to mitigate the toxicity of IL-12 by creating inactive split IL-12 subunits and assembling functional IL-12p70 predominantly in the tumor and tumor microenvironment.

Conclusions

- Increasing the interval of time between subunit injections or reducing the dose level of the second subunit effectively modulated serum drug concentration with minimal impact in the tumor.
- Inactive split IL-12p35 and IL-12p40 subunits assembled and formed functional IL-12p70 in non-human primates.
- Functional IL-12p70 complex caused activation and proliferation of target immune cell populations and induced cytokine and biomarker production.
- Tumor targeted self-assembling split IL-12 subunits represent a novel strategy to unlock the potential of IL-12p70 as a therapeutic by mitigating the toxicity associated with systemic administration.

Background

- Cytokines are a class of promising immunomodulatory proteins being explored as therapeutics[1].
- There has been limited translational success of the class due to their rapid clearance, pleiotropic properties, and associated toxicities[1].
- IL-12p70 is a heterodimeric cytokine comprised of p35 and p40 subunits[2].
- IL-12p70 is a potent stimulator of the immune system and can have profound anti-tumor activity, including IFNγ production, increased immune infiltration, enhanced NK and T cell activation, and reduced angiogenesis[3-4].
- The clinical use of IL-12p70 has been limited due to poor systemic tolerability[5].
- We sought to develop an engineered IL-12 receptor agonist which could be systemically administered and improve the therapeutic index.

Strategy

- Individual IL-12p35 or p40 subunits were separately fused to non-competitive antibody fragments targeting a tumor associated antigen that is highly expressed in a wide variety of malignancies.
- The goal of this approach was to preferentially assemble IL-12p70 complexes in the tumor microenvironment (TME) by sequential administration of the targeted subunits.
- Optimizing the dose level and interval of time between subunit administration will maintain tumor exposure while reducing systemic drug concentration and thereby potentially ameliorating associated toxicities with systemic IL-12 delivery (**Figure 1**).

Figure 1. Mural Oncology Approach to Expand Therapeutic Window with Systemic IL-12p70 Delivery

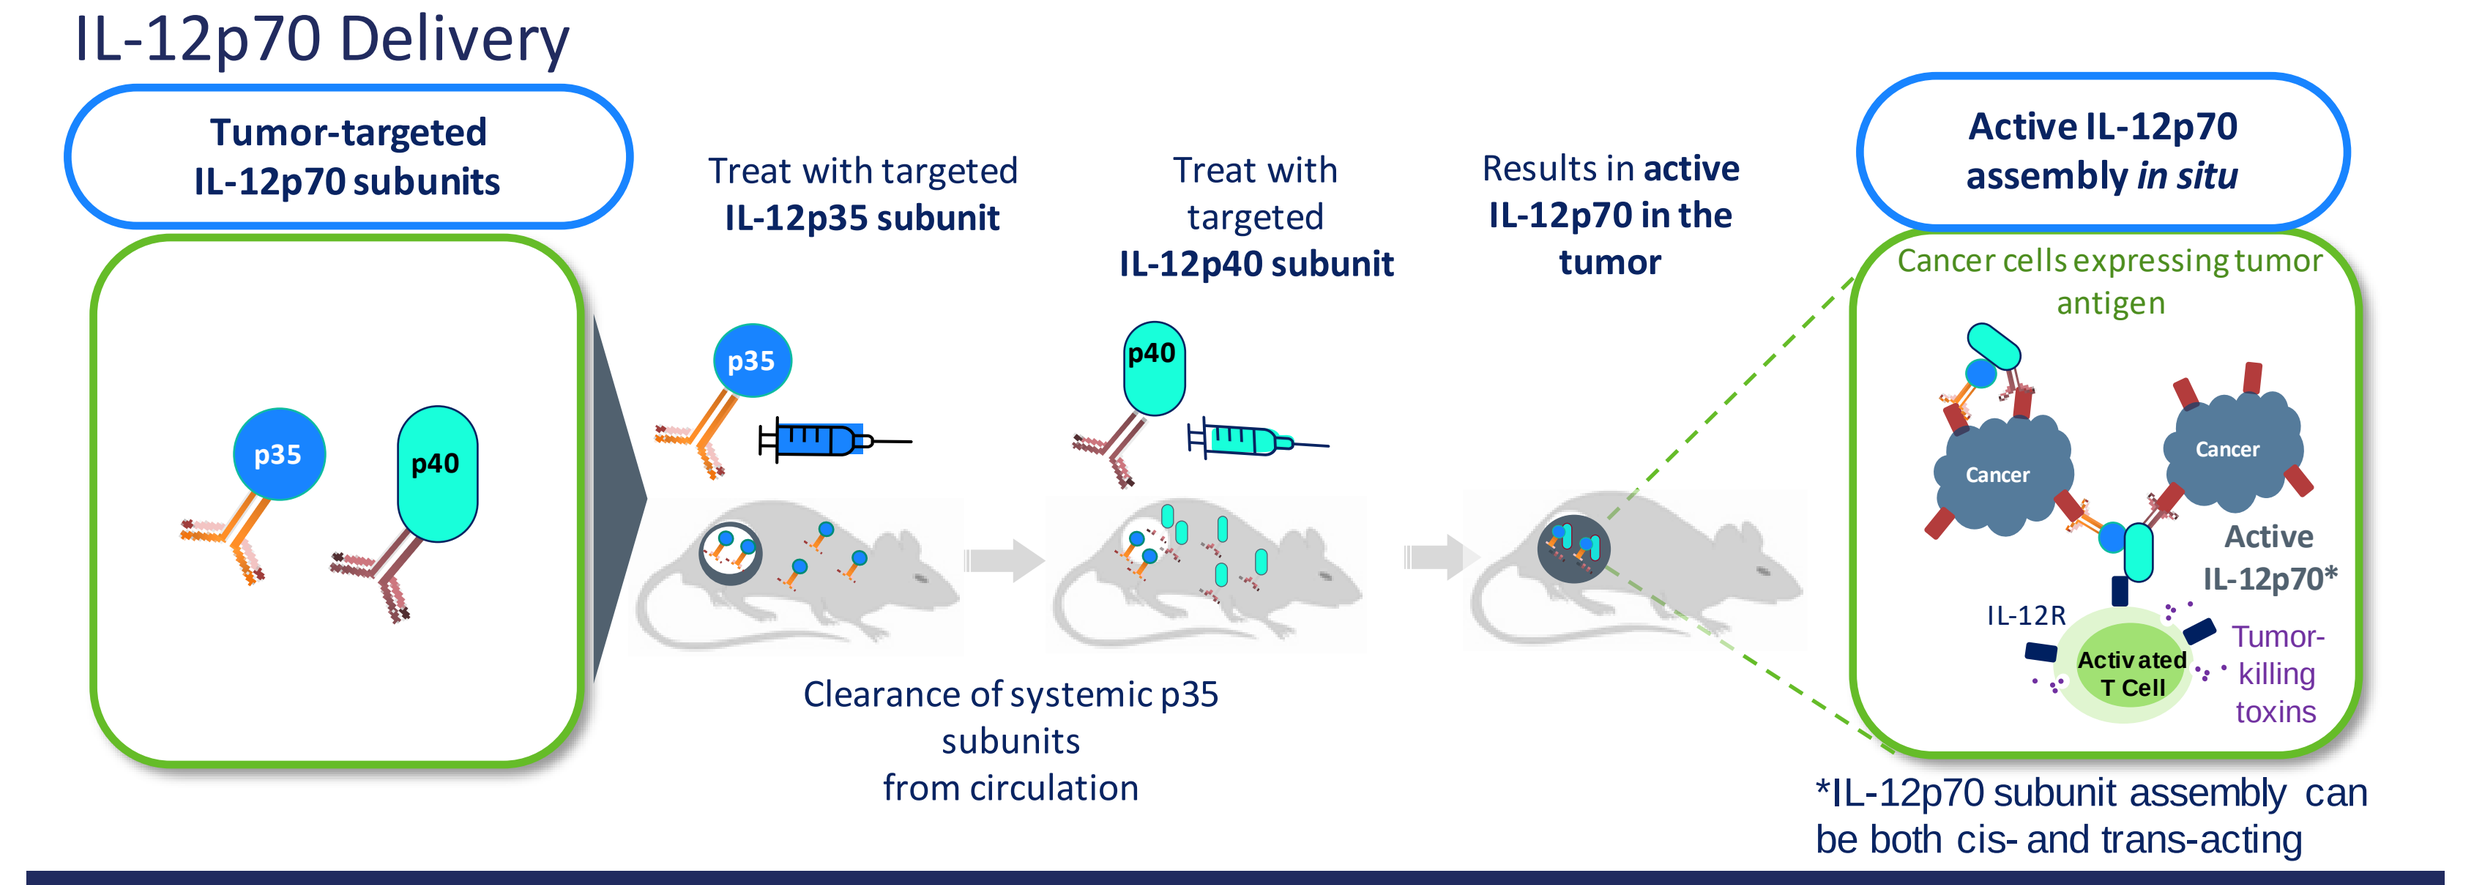
Tumor-targeted IL-12p70 subunits
p35
p40
Treat with targeted IL-12p35 subunit
Treat with targeted IL-12p40 subunit
Results in active IL-12p70 in the tumor
Clearance of systemic p35 subunits from circulation
Active IL-12p70 assembly in situ
Cancer cells expressing tumor antigen
Cancer
Active IL-12p70*
IL-12R
Activated T Cell
Tumor-killing toxins
*IL-12p70 subunit assembly can be both cis- and trans-acting

Methods

- Pharmacokinetics of individual IL-12p35 and IL-12p40 subunits and the resulting complex were evaluated in preclinical mouse models, including wild type BALB/c mice, tumor bearing immunocompromised mice, and tumor bearing humanized mice.
- Pharmacokinetics and pharmacodynamics were also assessed in non-human primates.
- Serum analytes were measured using plate-based MSD immunoassays and immunophenotyping was performed on a Cytek Aurora spectral cytometer.

Results

Figure 2. Increasing the Time Interval Between Subunit Administration Reduced IL-12p70 Exposure

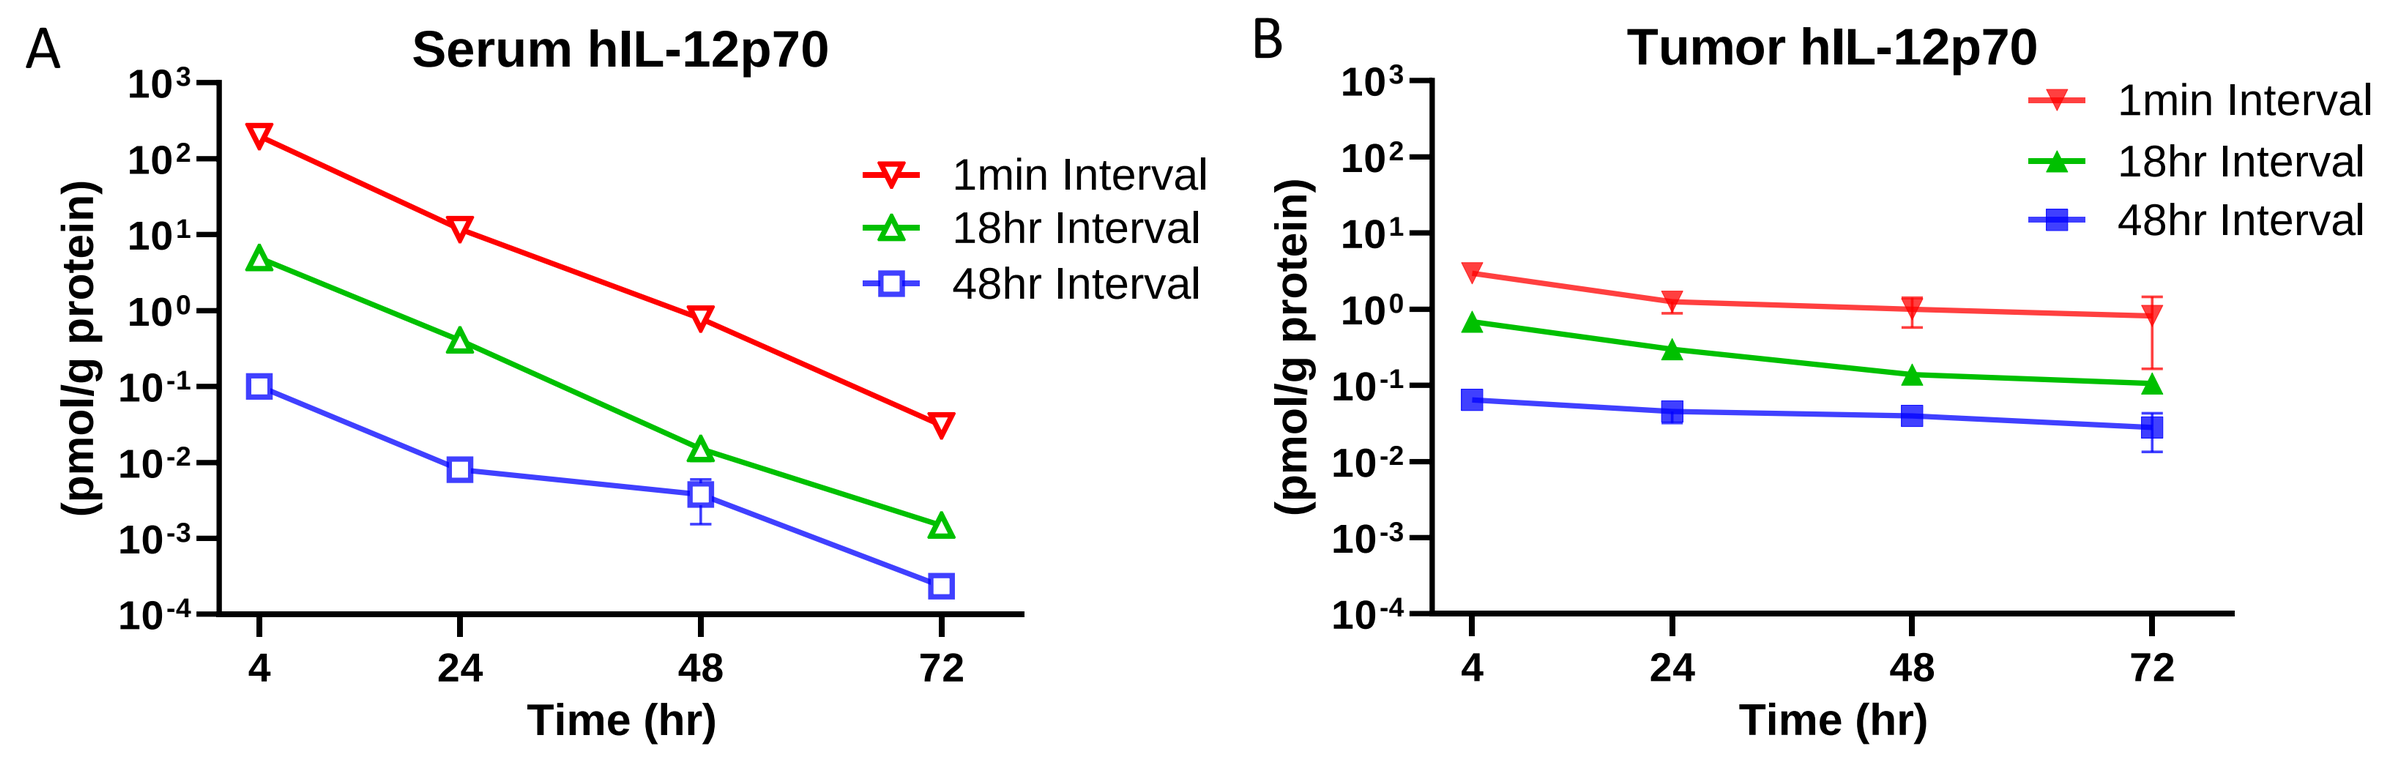
A
Serum hIL-12p70
B
Tumor hIL-12p70
(pmol/g protein)
Time (hr)
1min Interval
18hr Interval
48hr Interval

- NCG mice were engrafted with RKO (human colon carcinoma) xenografts and treated in sequence with split IL-12 subunits with increasing time interval between injections.
- The relative half-life increased dramatically in the tumor (vs. serum) demonstrating that targeting to the tumor associated antigen led to molecule retention in the tumor.
- Apparent C_{max} measured 4 hours after the second subunit was administered showed a significant reduction in serum compared to tumor as the interval increased.
- These pharmacokinetic profiles confirm the potential of the design hypothesis.

Results (continued)

- Female NOD/SCID mice bearing RKO (human colon carcinoma) xenografts (**Figures 3-5**)

Figure 3. Increasing the Interval Between Subunit Injections from 1 minute to 4 hours Reduced IL-12p70 in Serum but not in the Tumor

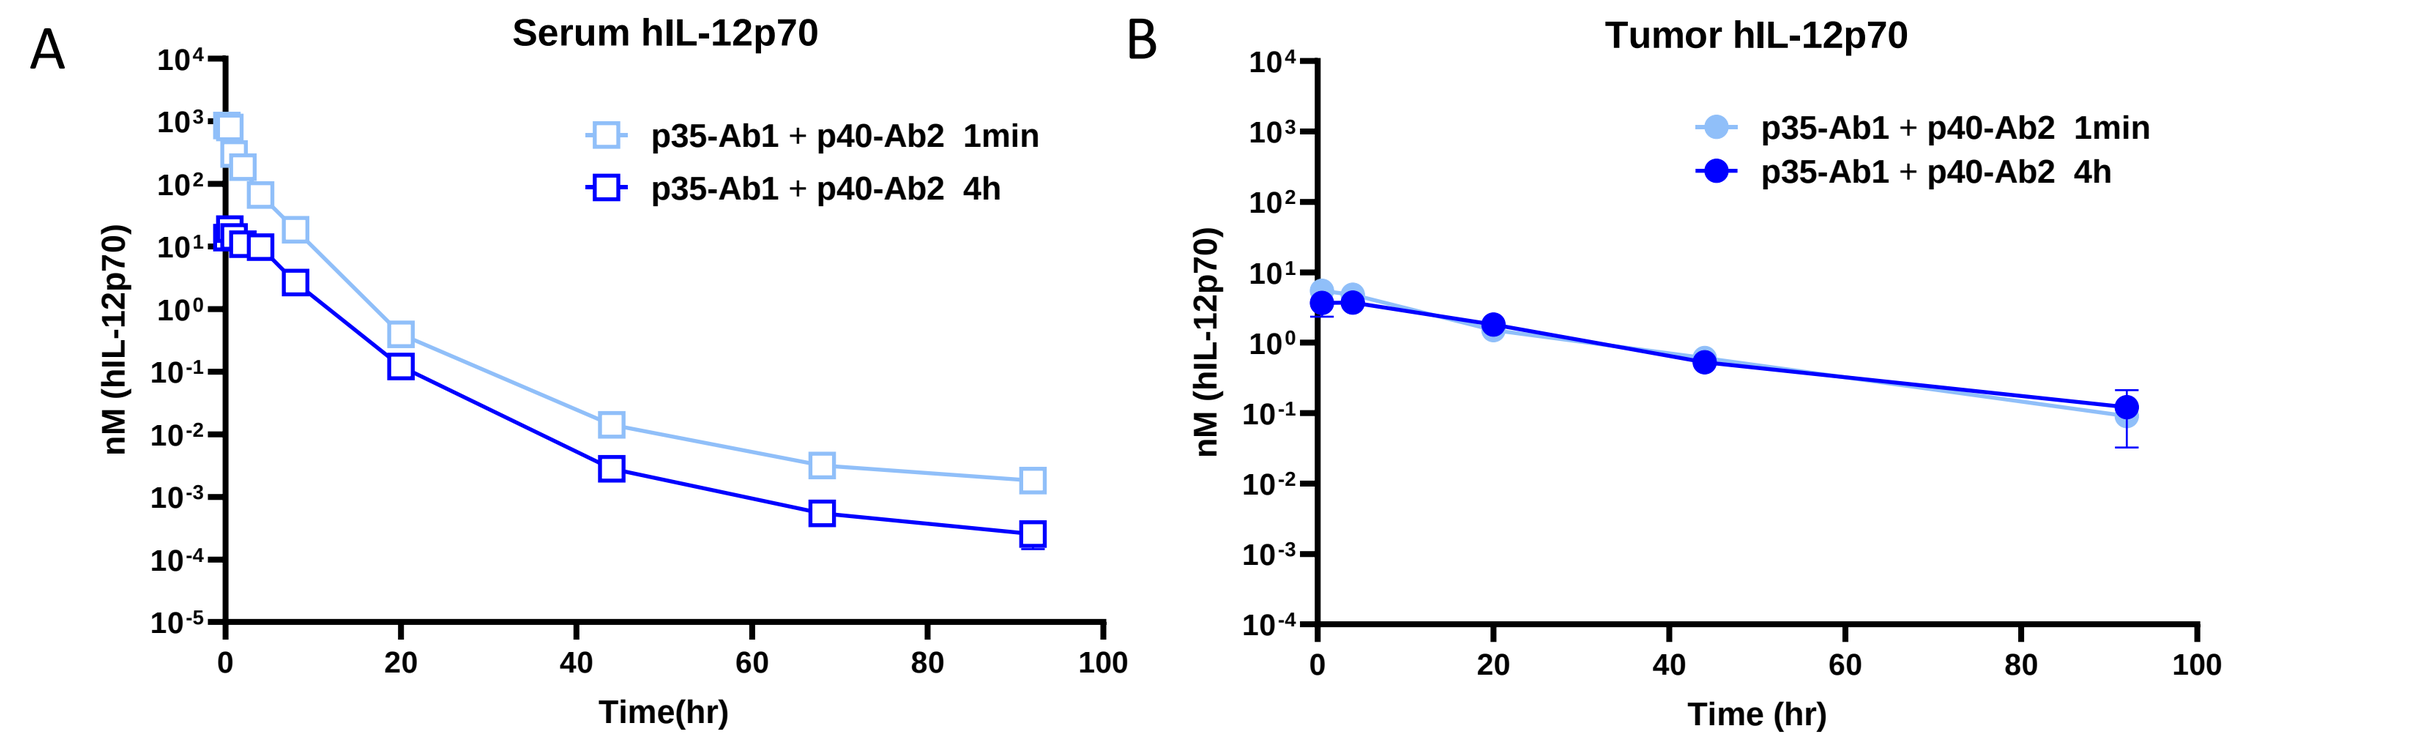
A
Serum hIL-12p70
p35-Ab1 + p40-Ab2 1min
p35-Ab1 + p40-Ab2 4h
nM (hIL-12p70)
Time(hr)
B
Tumor hIL-12p70
p35-Ab1 + p40-Ab2 1min
p35-Ab1 + p40-Ab2 4h
Time (hr)

- Extending the dosing interval between subunits from 1 minute to 4 hours led to a 45-fold reduction in C_{max} and a 25.4-fold reduction in AUC in the serum. (**Figure 3A)** C_{max} and AUC were nearly identical in the tumor (**Figure 3B**).
- After a 4-hour interval IL-12p70 levels in the tumor demonstrated a ~13-fold increase in $t_{1/2}$ and a 2-fold increase in AUC in the tumor compared to serum, showing enhanced retention of IL-12p70 in the tumor.
- Complexed IL-12p70 has a faster clearance from the serum compared to the tumor, and increasing the interval between subunit dosing reduces serum levels but maintains enhanced tumor retention of the targeted subunits.

Figure 4. Reducing the Dose Level of the Second p40 Subunit has a Significant Impact on IL-12p70 Assembly and Retention in the Tumor

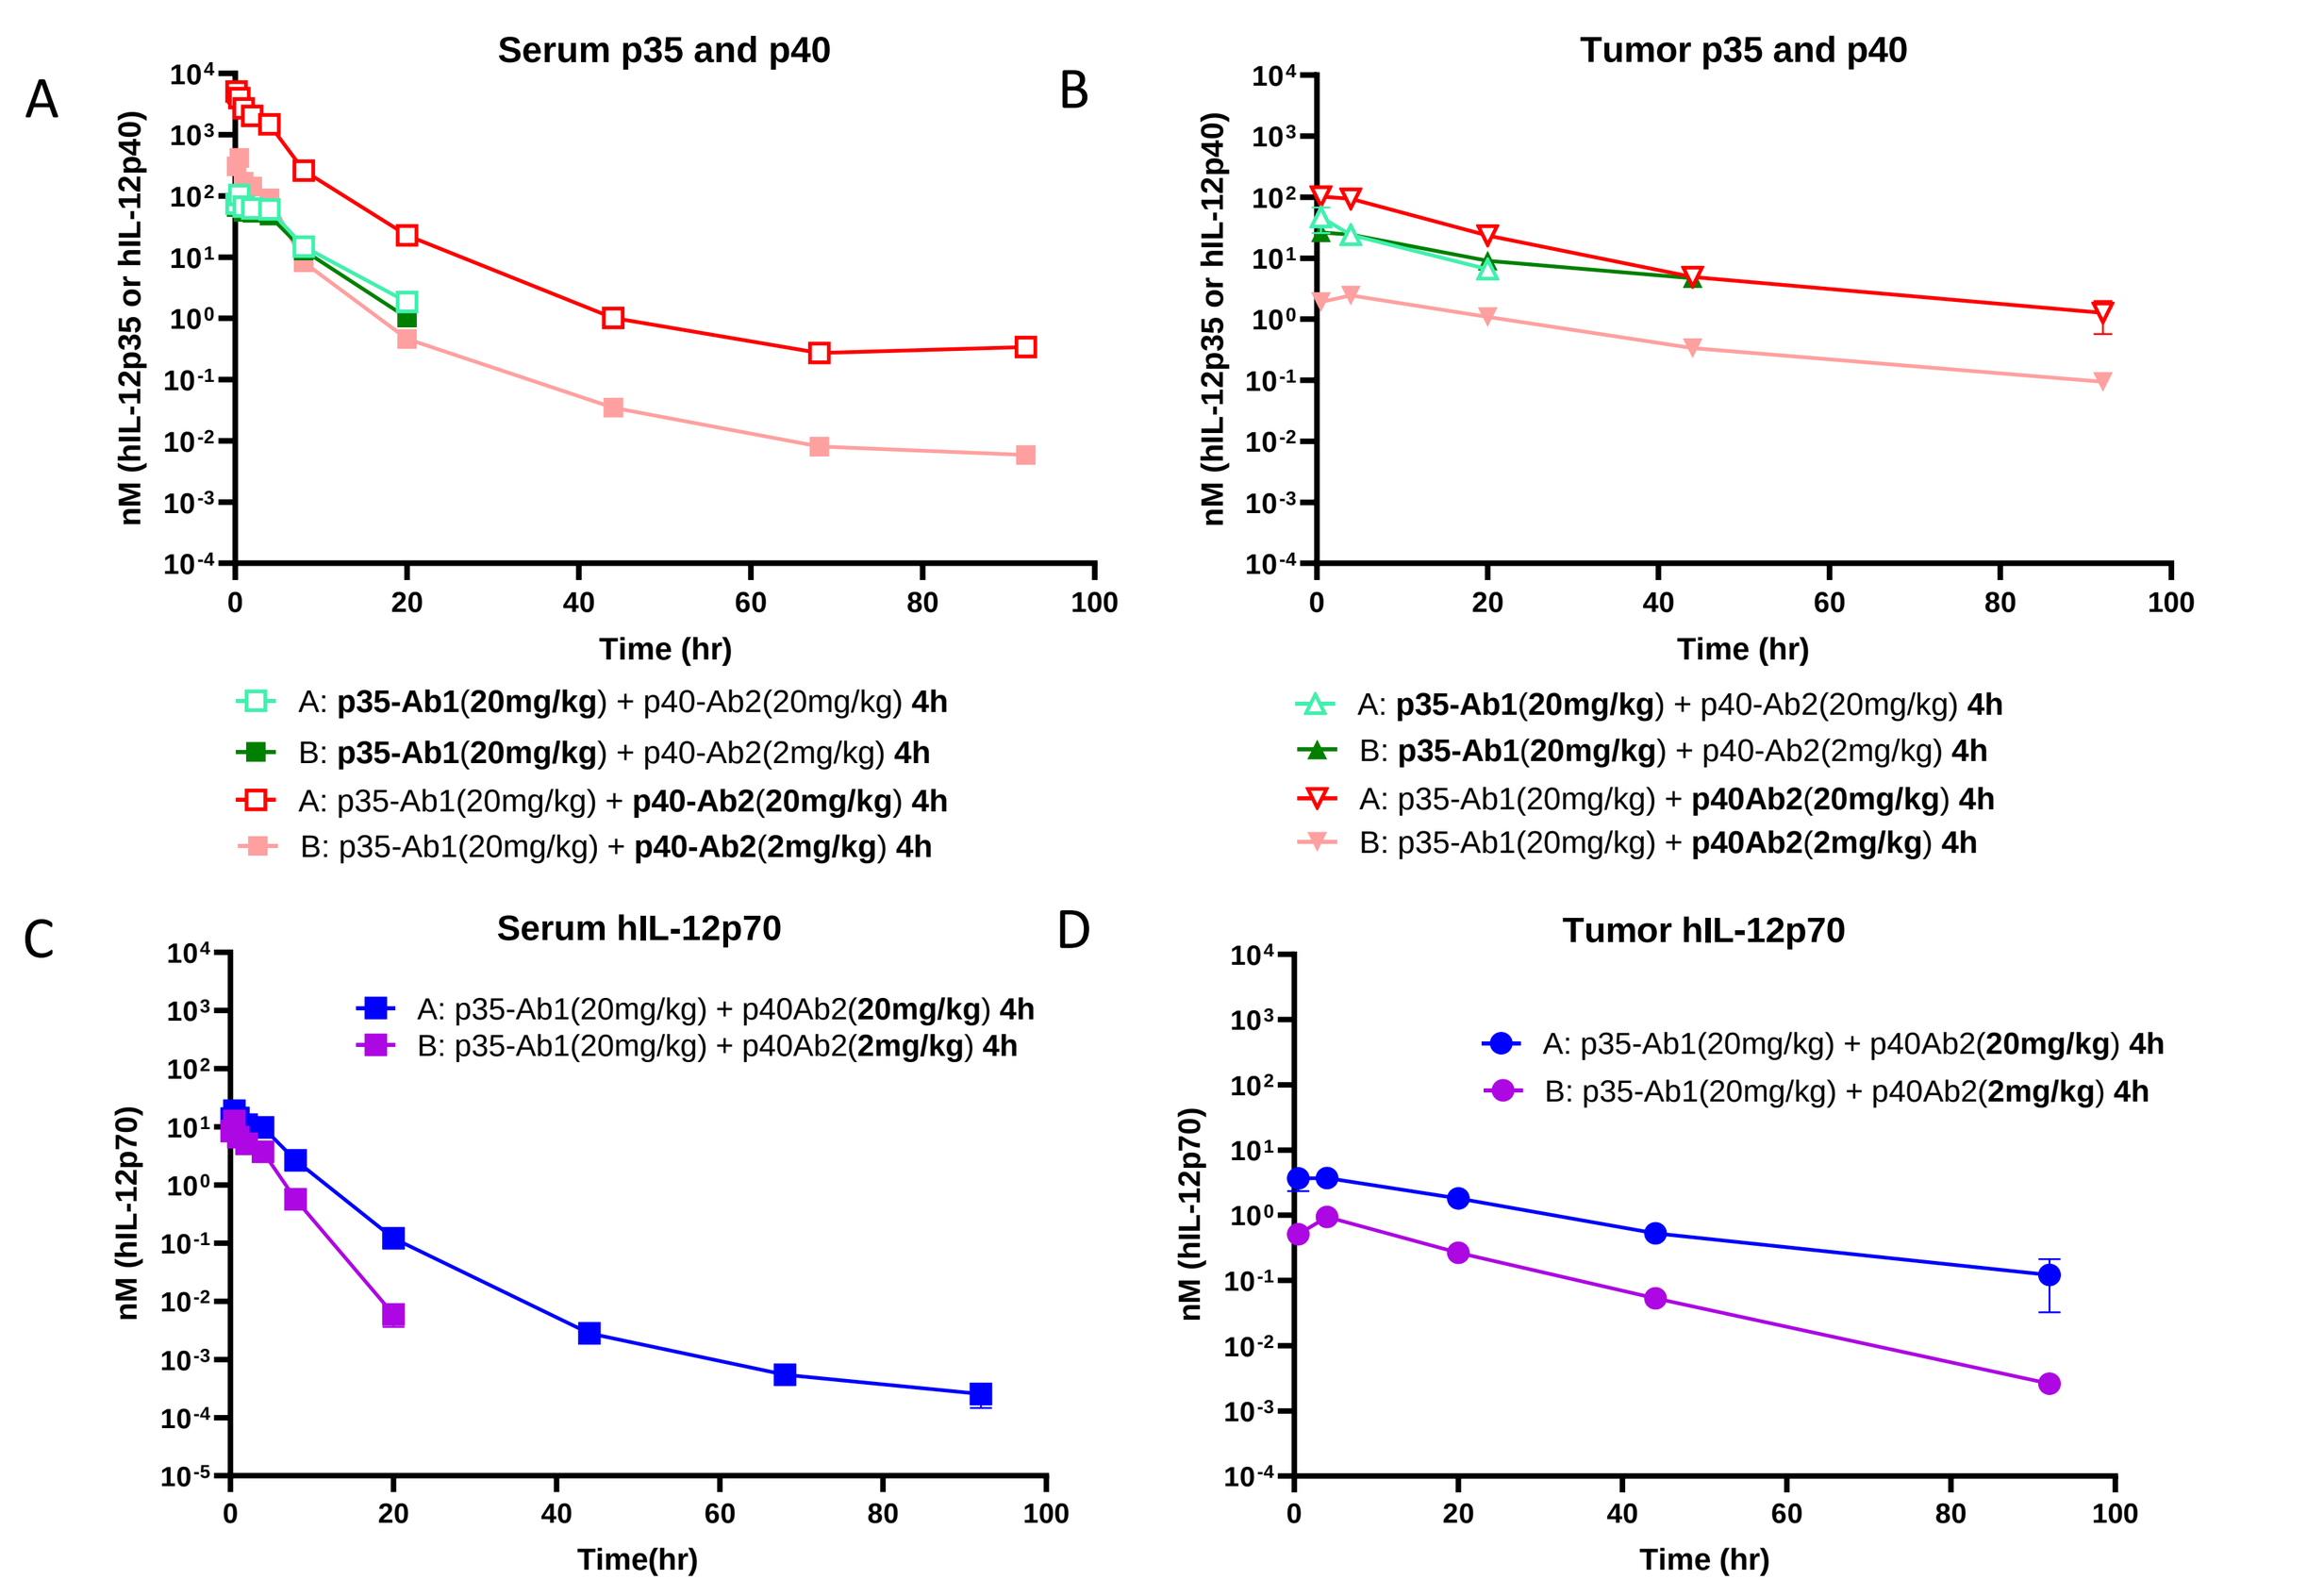
A
Serum p35 and p40
nM (hIL-12p35 or hIL-12p40)
Time (hr)
A: p35-Ab1(20mg/kg) + p40-Ab2(20mg/kg) 4h
B: p35-Ab1(20mg/kg) + p40-Ab2(2mg/kg) 4h
A: p35-Ab1(20mg/kg) + p40-Ab2(20mg/kg) 4h
B: p35-Ab1(20mg/kg) + p40-Ab2(2mg/kg) 4h
B
Tumor p35 and p40
A: p35-Ab1(20mg/kg) + p40-Ab2(20mg/kg) 4h
B: p35-Ab1(20mg/kg) + p40-Ab2(2mg/kg) 4h
A: p35-Ab1(20mg/kg) + p40Ab2(20mg/kg) 4h
B: p35-Ab1(20mg/kg) + p40Ab2(2mg/kg) 4h
C
Serum hIL-12p70
A: p35-Ab1(20mg/kg) + p40Ab2(20mg/kg) 4h
B: p35-Ab1(20mg/kg) + p40Ab2(2mg/kg) 4h
nM (hIL-12p70)
Time(hr)
D
Tumor hIL-12p70
A: p35-Ab1(20mg/kg) + p40Ab2(20mg/kg) 4h
B: p35-Ab1(20mg/kg) + p40Ab2(2mg/kg) 4h
Time (hr)

- All mice were administered the same dose level of p35. After a 4hr interval p40 dose was given at the same level as p35 or reduced 10-fold.
- 10-fold reduction of p40 resulted in a proportional decrease in serum p40 but greater than proportional reduction in tumor (**Figure 4A and B)**
- Reduction in p40 had minor impacts in serum but demonstrated a greater impact on p70 levels in the tumor (**Figure 4C and D**)
- These data show that although reducing the dose of the second subunit (p40) further reduced serum levels the greater impact on tumor p70 levels is contrary to the design hypothesis. Therefore, it is suggested to maximize the dose levels of both subunits with an appropriate interval selected to minimize serum and maintain tumor exposure.

Figure 5. Changing the PK of the Second Subunit Impacts IL-12p70 Assembly and Tumor Retention

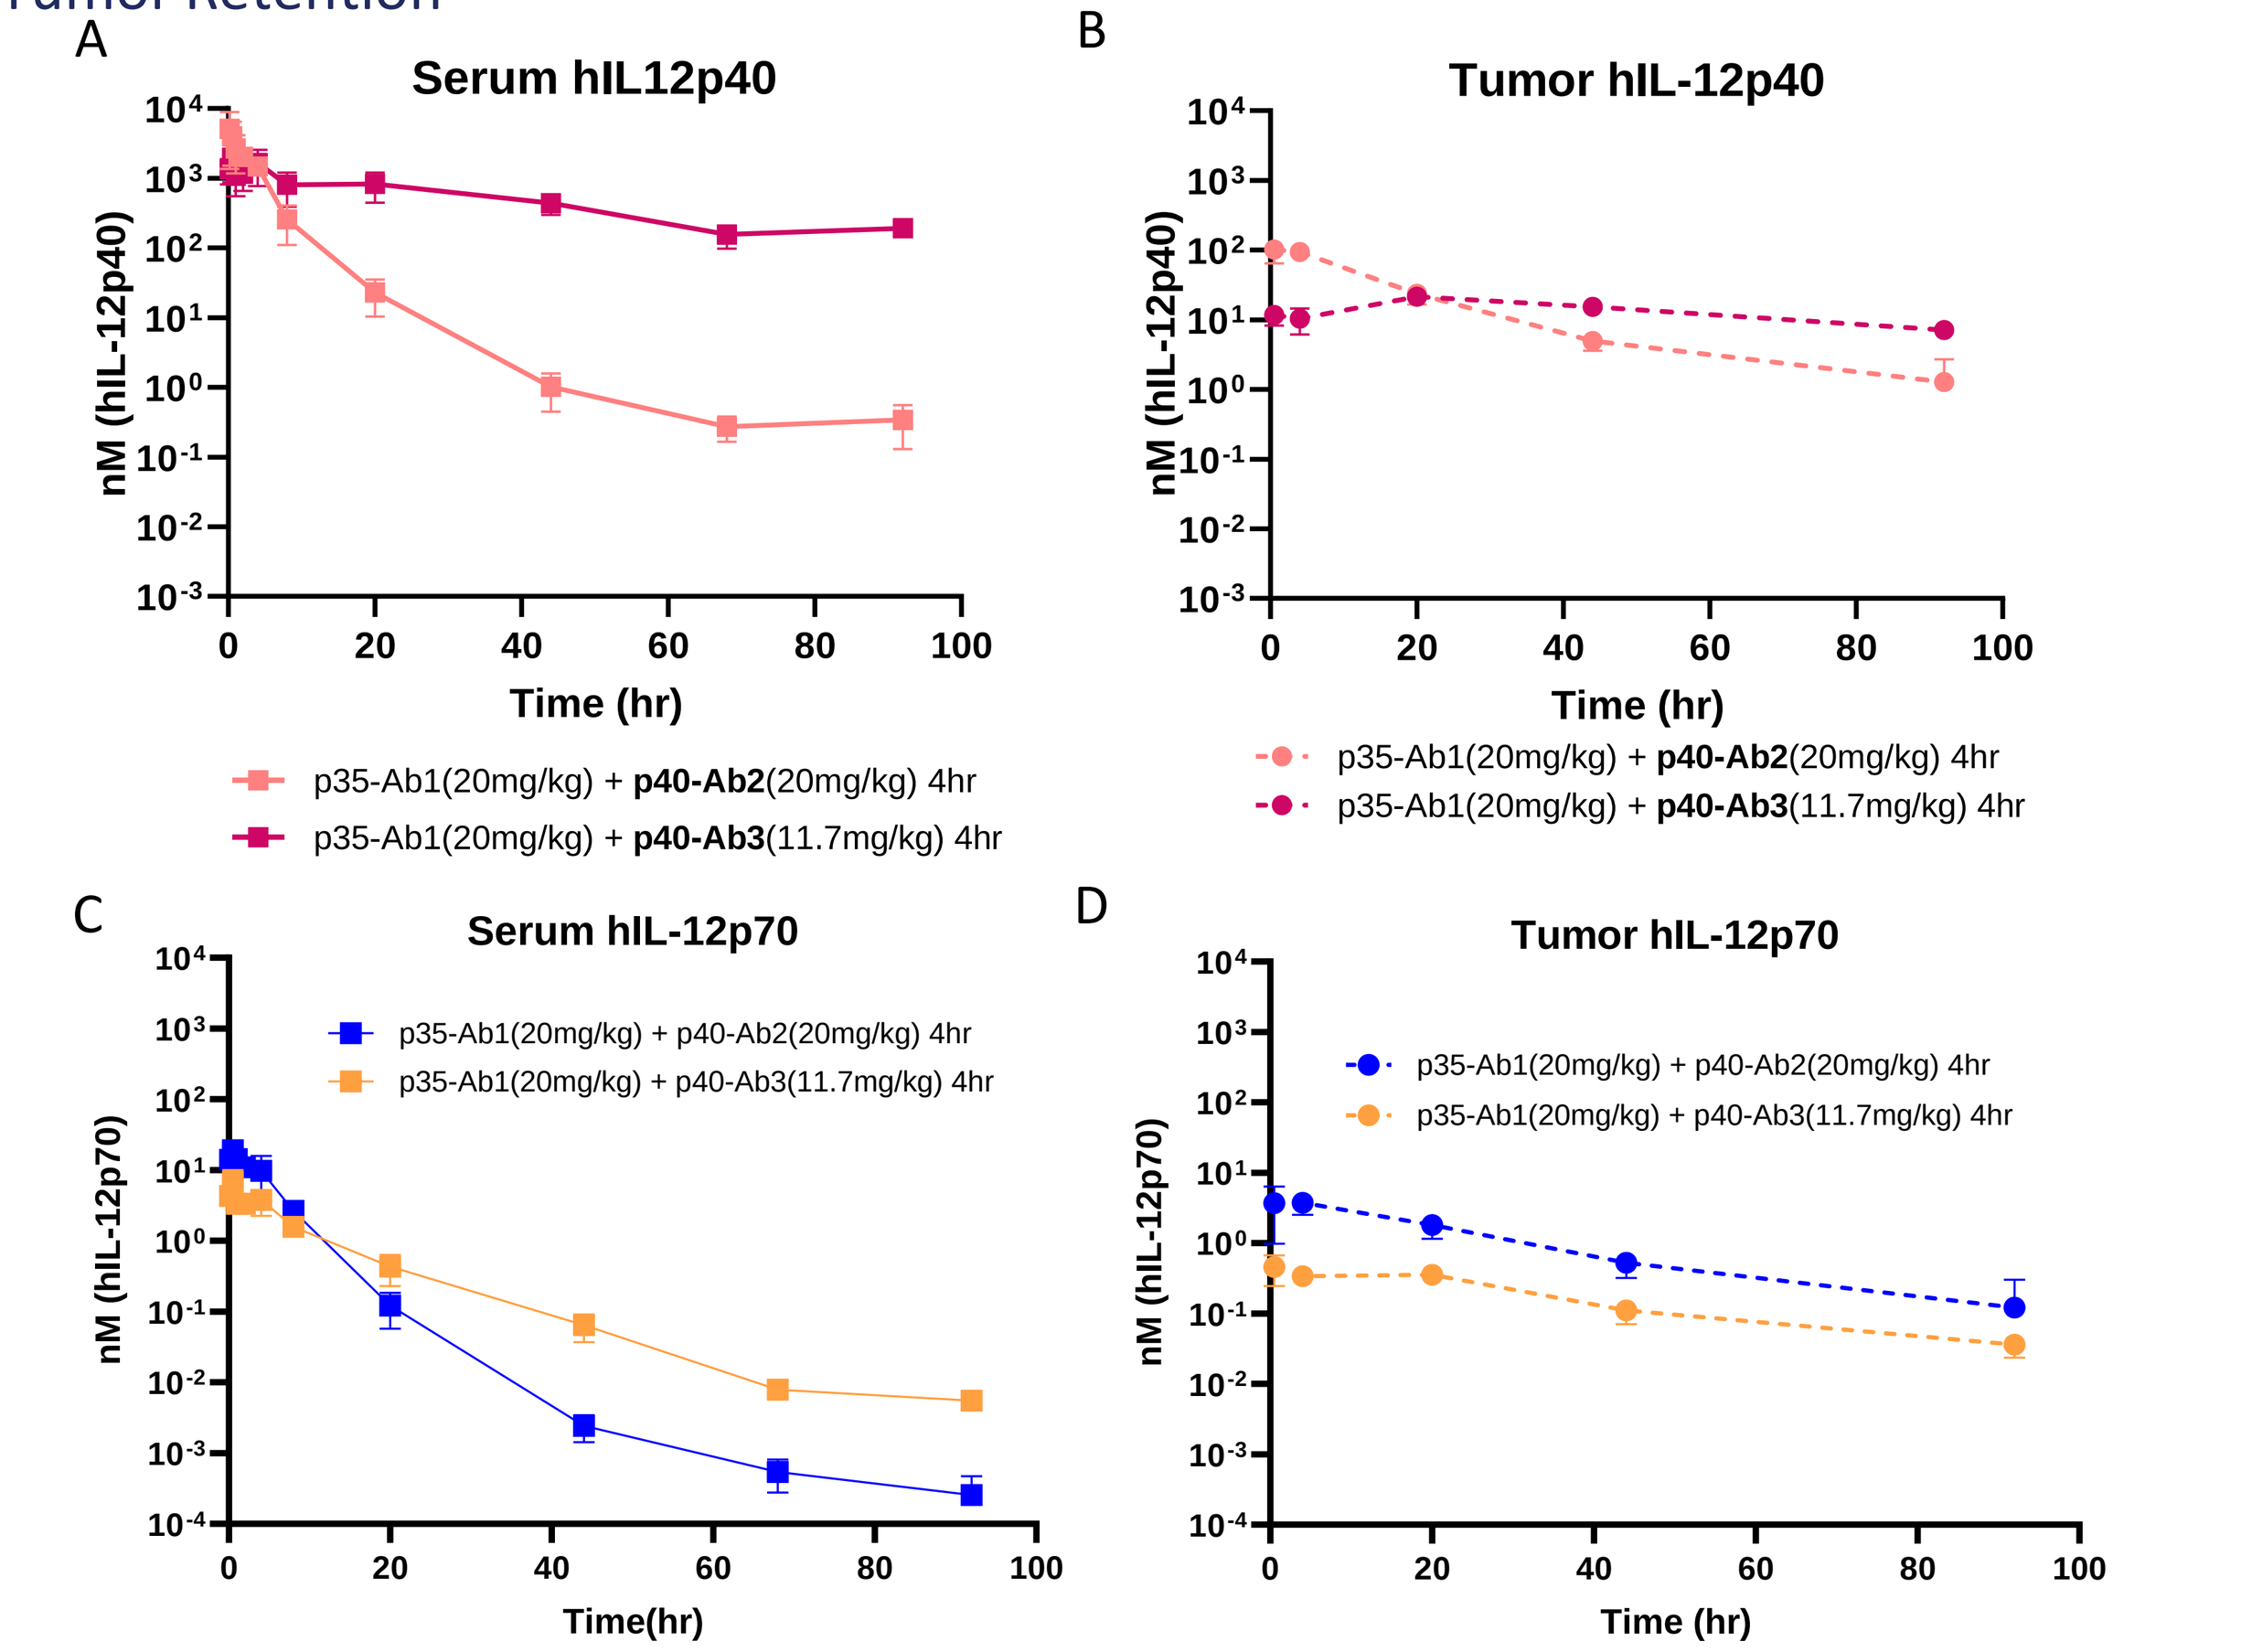
A
Serum hIL12p40
nM (hIL-12p40)
Time (hr)
p35-Ab1(20mg/kg) + p40-Ab2(20mg/kg) 4hr
p35-Ab1(20mg/kg) + p40-Ab3(11.7mg/kg) 4hr
B
Tumor hIL-12p40
p35-Ab1(20mg/kg) + p40-Ab2(20mg/kg) 4hr
p35-Ab1(20mg/kg) + p40-Ab3(11.7mg/kg) 4hr
C
Serum hIL-12p70
p35-Ab1(20mg/kg) + p40-Ab2(20mg/kg) 4hr
p35-Ab1(20mg/kg) + p40-Ab3(11.7mg/kg) 4hr
nM (hIL-12p70)
Time(hr)
D
Tumor hIL-12p70
p35-Ab1(20mg/kg) + p40-Ab2(20mg/kg) 4hr
p35-Ab1(20mg/kg) + p40-Ab3(11.7mg/kg) 4hr
Time (hr)

- Changing the fusion scaffold dramatically impacts subunit PK in serum and tumor
- In serum treatment with p40-Ab3 had reduced C_{max} (2.8-fold), similar AUC (1.3-fold difference), and increased $t_{1/2}$ (6.2-fold) compared to treatment with p40-Ab2 (**Figure 5A**)
- In tumor p40-Ab3 resulted in reduced C_{max} (8.2-fold) and exposure (6.3-fold) with a similar $t_{1/2}$ (1.3-fold difference) compared to p40-Ab2 (**Figure 5B**).
- Adjustments to PK of second subunit impact IL-12p70 assembly and retention in serum and tumor
- In serum complex formed with p40-Ab3 had reduced C_{max} and similar exposure compared to IL-12p70 complex formed with p40-Ab2 (**Figure 5C**)
- By contrast in tumor: IL-12p70 complex formed with p40-Ab3 had reduced C_{max} and exposure compared to complex formed with p40-Ab2 (**Figure 5D**)
- Using an IL-12p40 subunit fusion with smaller molecular size resulted in more rapid clearance of IL-12p70 from serum but greater accumulation/retention in the tumor

Figure 6. Pharmacokinetics from Mouse Translate to NHP

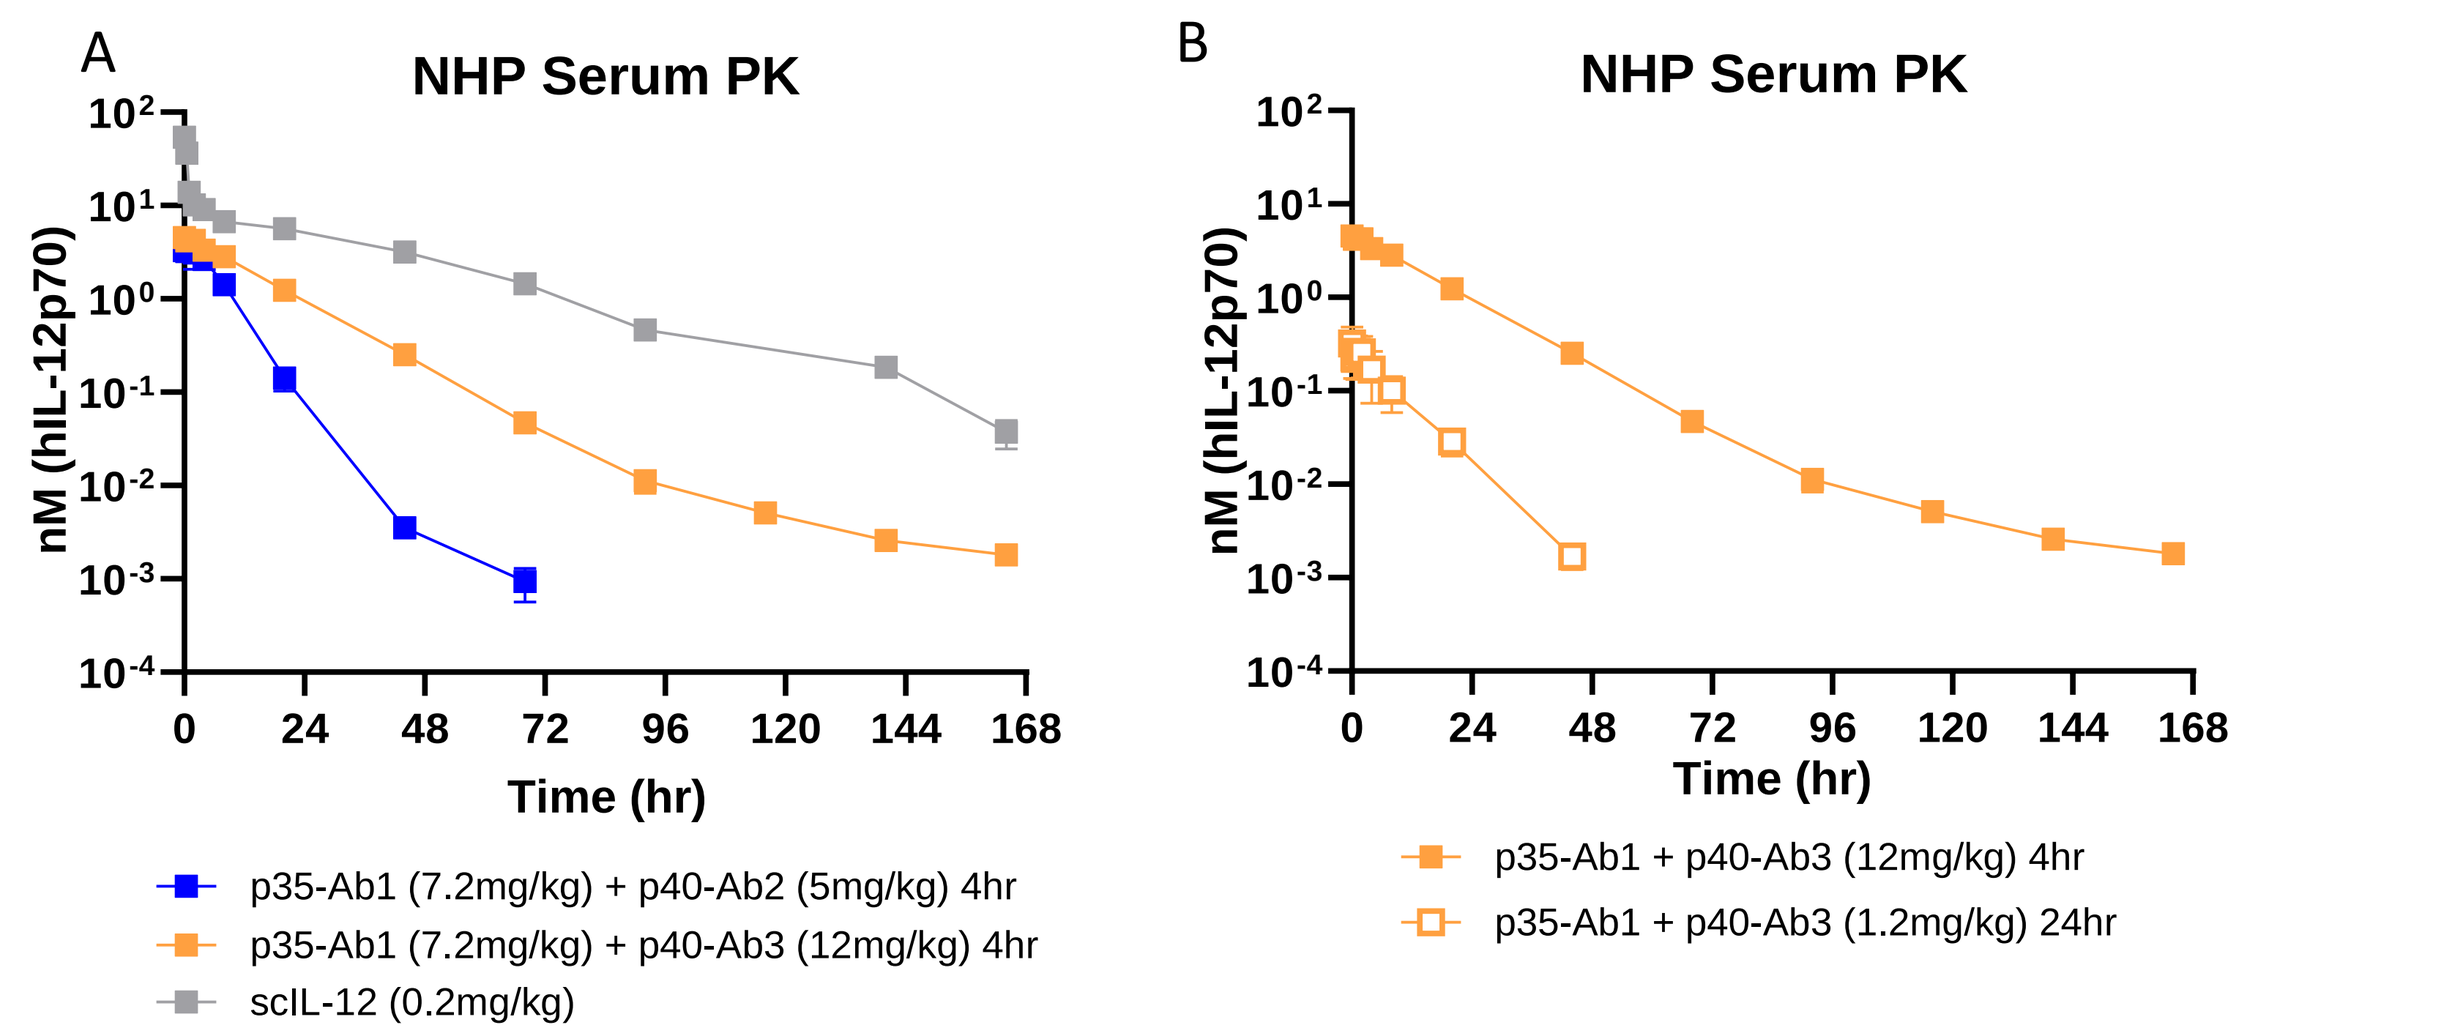
A
NHP Serum PK
nM (hIL-12p70)
Time (hr)
p35-Ab1 (7.2mg/kg) + p40-Ab2 (5mg/kg) 4hr
p35-Ab1 (7.2mg/kg) + p40-Ab3 (12mg/kg) 4hr
scIL-12 (0.2mg/kg)
B
NHP Serum PK
p35-Ab1 + p40-Ab3 (12mg/kg) 4hr
p35-Ab1 + p40-Ab3 (1.2mg/kg) 24hr

- n=5/group naïve NHP were treated with p35-Ab1 and either p40-Ab2 or p40-Ab3 after a 4- or 24-hour interval between subunit injections.
- The changes in IL-12p70 PK parameters (C_{max}, AUC, $t_{1/2}$) observed in NHP is similar to that observed in mice when the PK characteristics of the second subunit change (**Figure 6A**).
- Additionally, increasing the interval of time between injections while reducing the dose of the second subunit resulted in a decrease in exposure (**Figure 6B**).
- Note that the dose selected for scIL-12 resulted in a higher exposure compared to split IL-12 subunits. This was included to control for IL-12 pharmacodynamic activity (**Figure 7**).

Figure 7. Split IL-12 Subunits are Active in NHP

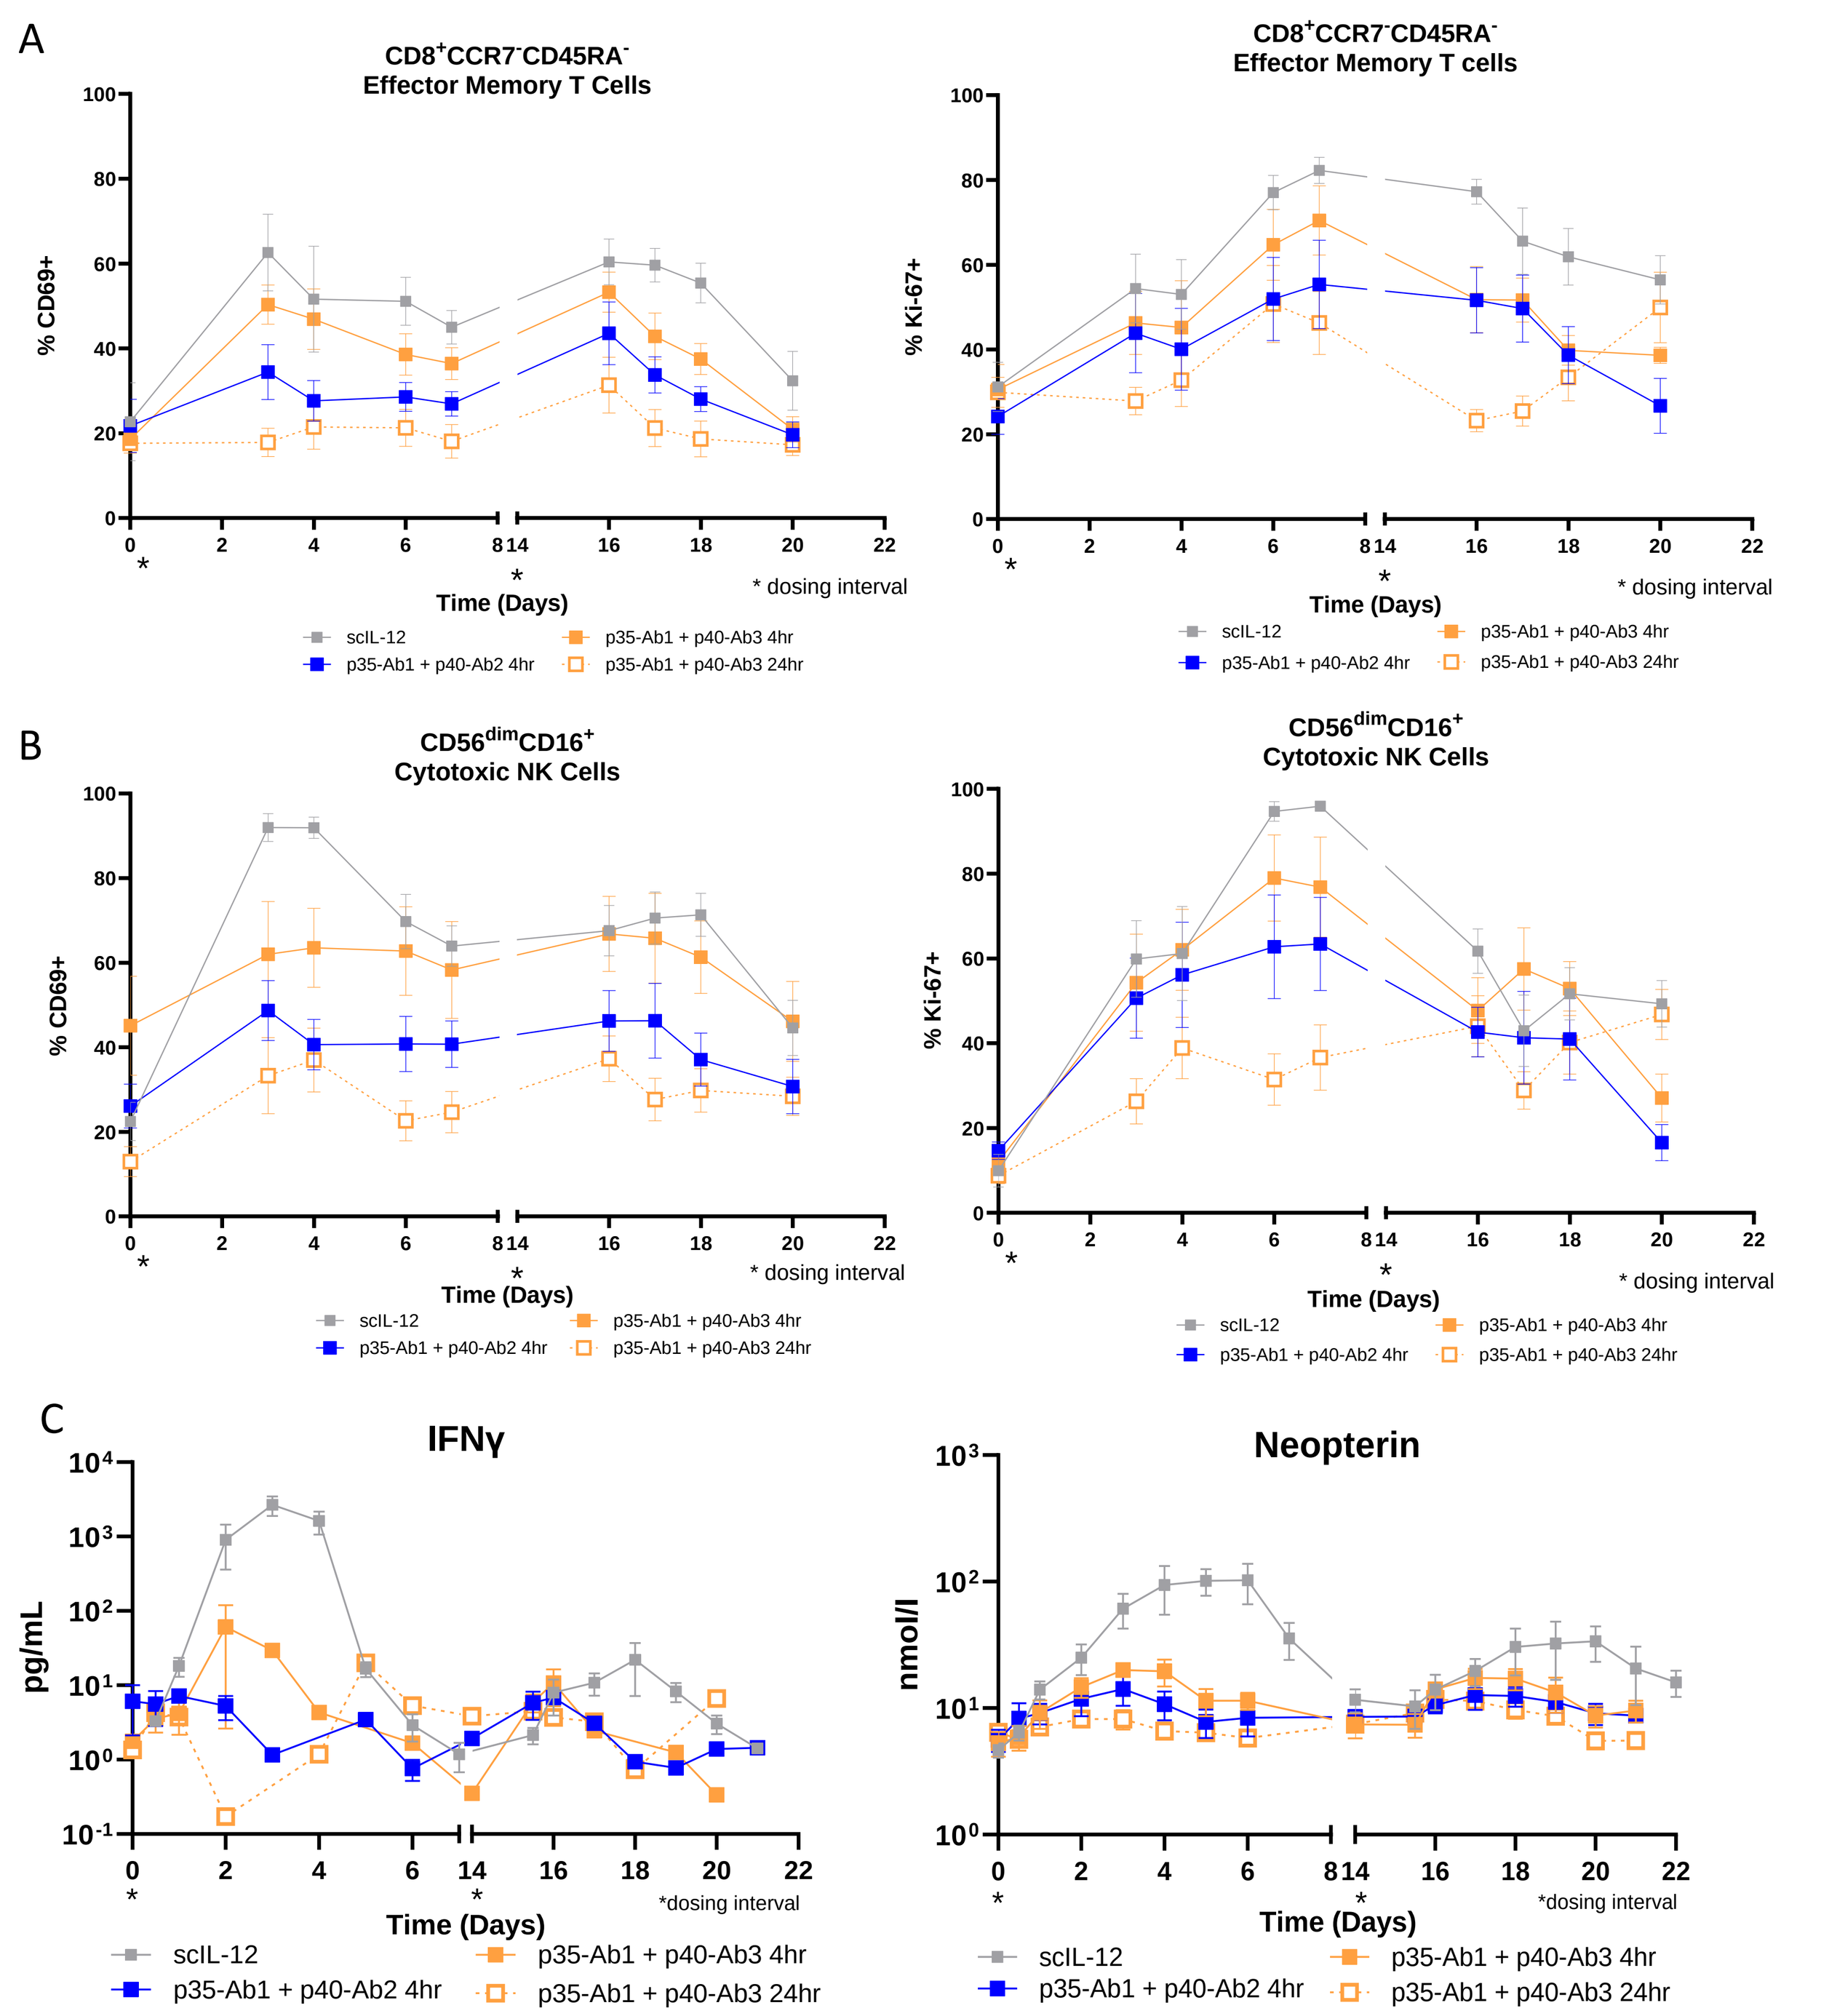
A
CD8+CCR7-CD45RA- Effector Memory T Cells
% CD69+
CD8+CCR7-CD45RA- Effector Memory T cells
% Ki-67+
Time (Days)
* dosing interval
B
CD56dimCD16+ Cytotoxic NK Cells
% CD69+
CD56dimCD16+ Cytotoxic NK Cells
% Ki-67+
C
IFNγ
pg/mL
Neopterin
nmol/l
scIL-12
p35-Ab1 + p40-Ab2 4hr
p35-Ab1 + p40-Ab3 4hr
p35-Ab1 + p40-Ab3 24hr

- Blood CD8⁺ effector memory T cells (CCR7⁻ CD45RA⁻) and cytotoxic NK cells (CD56^dim CD16⁺) are activated (CD69⁺) and proliferating (Ki-67⁺) in an IL-12 exposure dependent manner (**Figure 7A and B**).
- Biomarkers of IL-12 response, IFNγ and Neopterin also showed a dose response relationship. Treatments with the highest exposure elicited the strongest cytokine/biomarker response (**Figure7C**).
- Changing the PK characteristics (AUC and $t_{1/2}$) of the second subunit or increasing the interval between subunits resulted in a modulation in overall cellular and biomarker responses.

References

1. Propper DJ, Balkwill FR. Harnessing cytokines and chemokines for cancer therapy. Nat Rev Clin Oncol. 2022 Apr;19(4):237-253. doi: 10.1038/s41571-021-00588-9. Epub 2022 Jan 7. PMID: 34997230.
2. Aragane Y, Riemann H, Bhardwaj RS, Schwarz A, Sawada Y, Yamada H, Luger TA, Kubin M, Trinchieri G, Schwarz T. IL-12 is expressed and released by human keratinocytes and epidermoid carcinoma cell lines. J Immunol. 1994 Dec 15;153(12):5366-72. PMID: 7527439.
3. Lasek W, Zagożdżon R, Jakobisiak M. Interleukin 12: still a promising candidate for tumor immunotherapy? Cancer Immunol Immunother. 2014 May;63(5):419-35. doi: 10.1007/s00262-014-1523-1. Epub 2014 Feb 11. PMID: 24514955; PMCID: PMC3994286.
4. Greiner JW, Morillon YM 2nd, Schlom J. NHS-IL12, a Tumor-Targeting Immunocytokine. Immunotargets Ther. 2021 May 27;10:155-169. doi: 10.2147/ITT.S306150. PMID: 34079772; PMCID: PMC8166332.
5. Leonard JP, Sherman ML, Fisher GL, Buchanan LJ, Larsen G, Atkins MB, Sosman JA, Dutcher JP, Vogelzang NJ, Ryan JL. Effects of single-dose interleukin-12 exposure on interleukin-12-associated toxicity and interferon-gamma production. Blood. 1997 Oct 1;90(7):2541-8. PMID: 9326219.

Acknowledgments

- We would like to thank all the current and former team members that have contributed to this project. Without you none of this would be possible.

SITC 39th Annual Meeting, Houston, TX • November 6-10, 2024