Preclinical Efficacy and Immune Activity of Half-life Extended IL-18 Fusion Proteins Resistant to IL-18BP Suppression.

Mark Whitmore,[1] Kristiana Dreaden,[1] Pinar Gurel,[1] Joshua Heiber,[1] Robert G Newman,[1] Su-Ping Pearson,[1] Chunhua Wang,[1] Yanchun Zhao[1] and Jean Chamoun[1]

[1]Mural Oncology Inc., 852 Winter Street, Waltham, MA 02451

Take Away

- Mural Oncology has developed IL-18BP resistant, half-life extended variants of IL-18 that demonstrate good safety and efficacy in preclinical models.
- These attributes are potentially transformative in developing a best-in-class immune-oncology therapeutic.

Conclusions

- In preclinical studies, Mural Oncology IL-18BP-resistant, half-life enhanced IL-18 variants overcome limitations of recombinant wild-type IL-18 for immune-oncology.
- The weekly (q7d) dosing regimen in mice provided durable immune responses and monotherapy efficacy holding promise that a transformative best-in-class immune-oncology therapeutic can be delivered with patient-friendly dosing.
- The safety margin of the mouse ortholog in dose-escalation efficacy studies suggest that monotherapy efficacy can be achieved with acceptable tolerability.
- These studies justify pursuit of IND-enabling preclinical studies for first-in-human clinical trials.

Background

- Human variants of Interleukin-18 (IL-18) with resistance to IL-18 binding protein (IL-18BP) suppression have been fused to Fragment Crystallizable (Fc) half-life enhancing protein scaffolds to generate molecules with optimal properties for a best-in-class immune-oncology therapeutic (Table 1).[1]
- Mural Oncology Human Variants of IL-18 fused to a human Fc scaffold (MhV-Fc) as well as a mouse ortholog of a human variant fused to a functionally equivalent mouse Fc scaffold (mO-Fc) show enhanced in vivo half-lives relative to rapidly-cleared naked tool variants of IL-18 with resistance to IL-18BP (mTV^{res}) (Table 1)[1].
- In longitudinal pharmacodynamic studies, half-life enhanced variants stimulated a serum T_H1-dominant cytokine increase of longer duration than naked mTV^{res}.[1]
- **In these studies, we show that Mural human IL-18BP-resistant half-life enhanced variants:**
 1. *Displayed in vitro anti-tumor activity by stimulating PBMC-mediated tumor growth inhibition of A549 cells in co-cultures with human PBMCs.*
 2. *Stimulated IFN-γ release from cynomolgus macaque PBMCs with potency that was comparable to human PBMCs.*
- **A mouse IL-18BP-resistant, half-life enhanced ortholog of a lead Mural human variant:**
 1. *Induced a more durable increase in blood cytotoxic NK and cytotoxic effector CD8⁺ T cells, two key immune mediators of anti-tumor activity, than short half-life mTV^{res}.*
 2. *Exhibited monotherapy efficacy in the heterotopic MC38 syngenic model of colorectal cancer with strong tumor growth inhibition, improved survival, complete responses, and a promising safety margin.*

Methods

In vitro biological activity of Mural hV-Fc

- *A549 growth inhibition in co-cultures of A549 cells with human PBMCs*. Nuclight Green transfected A549 cells were monitored for growth over 120h by IncuCyte live cell imaging in the presence and absence of human PBMCs. Concentration-response for MhV1-Fc and WT hIL-18 was evaluated based on viable A549 cells at 120h and normalized to viable cells cultured in the presence of PBMCs with no IL-18.
- *IFN-γ release assay from cynomolgus macaque PBMCs*. Human PBMCs were cultured in the presence and absence of increasing concentrations (as indicated) of MhV1-, MhV2-, or MhV3-Fc fusions as well as recombinant WT Rhesus IL-18 or WT human IL-18. Culture supernatants were collected at 24h and assessed for NHP IFN-γ by ELISA.

In vivo immune-biology and efficacy of mO-Fc

- *Longitudinal immune profiling of mouse blood immunocytes*. Non-tumor bearing C57BL/6 mice were treated by subcutaneous injection with the indicated doses of mO-Fc or mTV^{res}. Blood was collected at the indicated time points post-dose and stained for surface and intracellular antigens using a cocktail of fluorescently labeled antibodies. Expression was measured by multi-parameter flow cytometry and analyzed using FlowJo software.
- *Heterotopic MC38 syngeneic model of colorectal cancer*. MC38 cells were injected subcutaneously on the flank of C57BL/6 mice. Tumor volume (by caliper measurement) was monitored until tumors reached a mean volume if 80-120 mm³. Mice were randomized into treatment groups with equal distribution of tumor volume on Day 0. On days 1, 8, and 15 mice were injected subcutaneously at a site distal to the tumor with the indicated IL-18 molecule or vehicle (PBS). Tumor volume, body weight, and condition were monitored at the indicated time points over the duration of study.

Table 1. *In vitro properties (IL-18BP sensitivity and potency) and in vivo half-life of Mural Oncology human variants, mouse orthologs, and tool variants*. Half-discs represent IL-18 moiety and stacked ovals represent Fc half-life enhancing protein scaffolds.

Description (Abbreviation)	[1]Resistance to IL-18BP?	[2]Potency (EC_{50} - pM)	Half-life (h)
Mural Human Variant 1-hFc (MhV1-Fc)	Yes	4	14
Mural Human Variant 2-hFc (MhV2-Fc)	Yes	30	15
"Naked" Mouse Tool Variant^{resistant} (mTV^{res})	Yes	0.2	<0.6
Mouse Tool Variant^{sensitive}-Fc (mTV^{sens}-Fc)	No (sensitive)	250	ND
Mural Mouse Ortholog-Fc (mO-Fc)	Yes	1,278	30

Potency of WT Human IL-18 (EC_{50}): 307 ± 137 pM (Mean ± SEM) *Potency of WT Mouse IL-18 (EC_{50}): 14 ± 6 pM (Mean ± SEM)*

[1]Resistance to super-physiological IL-18BP concentration (300 nM) in PBMC IFN-γ release assay (splenocyte IFN-γ release in mice)
[2]Potency in PBMC IFN-γ release assay (splenocyte IFN-γ release in mice)
Pharmacokinetics of human variants tested in humanized mice

Results

Mural Human Variants stimulated tumor cell growth inhibition and are active on cynomolgus macaque PBMCs

- Mural hV1-Fc and WT Human IL-18 stimulated concentration-dependent tumor growth inhibition of A549 cells in co-cultures with human PBMCs (live cell imaging). Tumor cell growth inhibition was dependent on the presence of PBMCs.

Figure 1. *Mural hV1-Fc stimulated PBMC-mediated A549 tumor cell growth inhibition (A) and activated Cynomolgus PBMCs (B) with potency similar to that observed with human PBMCs. (see Table 1).* (A) IncuCyte live cell imaging of A549 cells in co-culture with human PBMC for 120h. Viable A549 cells at 120h relative to A549 + PBMCs (no IL-18 – dotted line) was as indicated for Mural hV1-Fc and WT hIL-18. (B) Concentration-response for stimulating IFN-γ release from cynomolgus macaque PBMCs. Wild-type Rhesus IL-18, WT hIL-18, MhV1-Fc and MhV2-Fc.

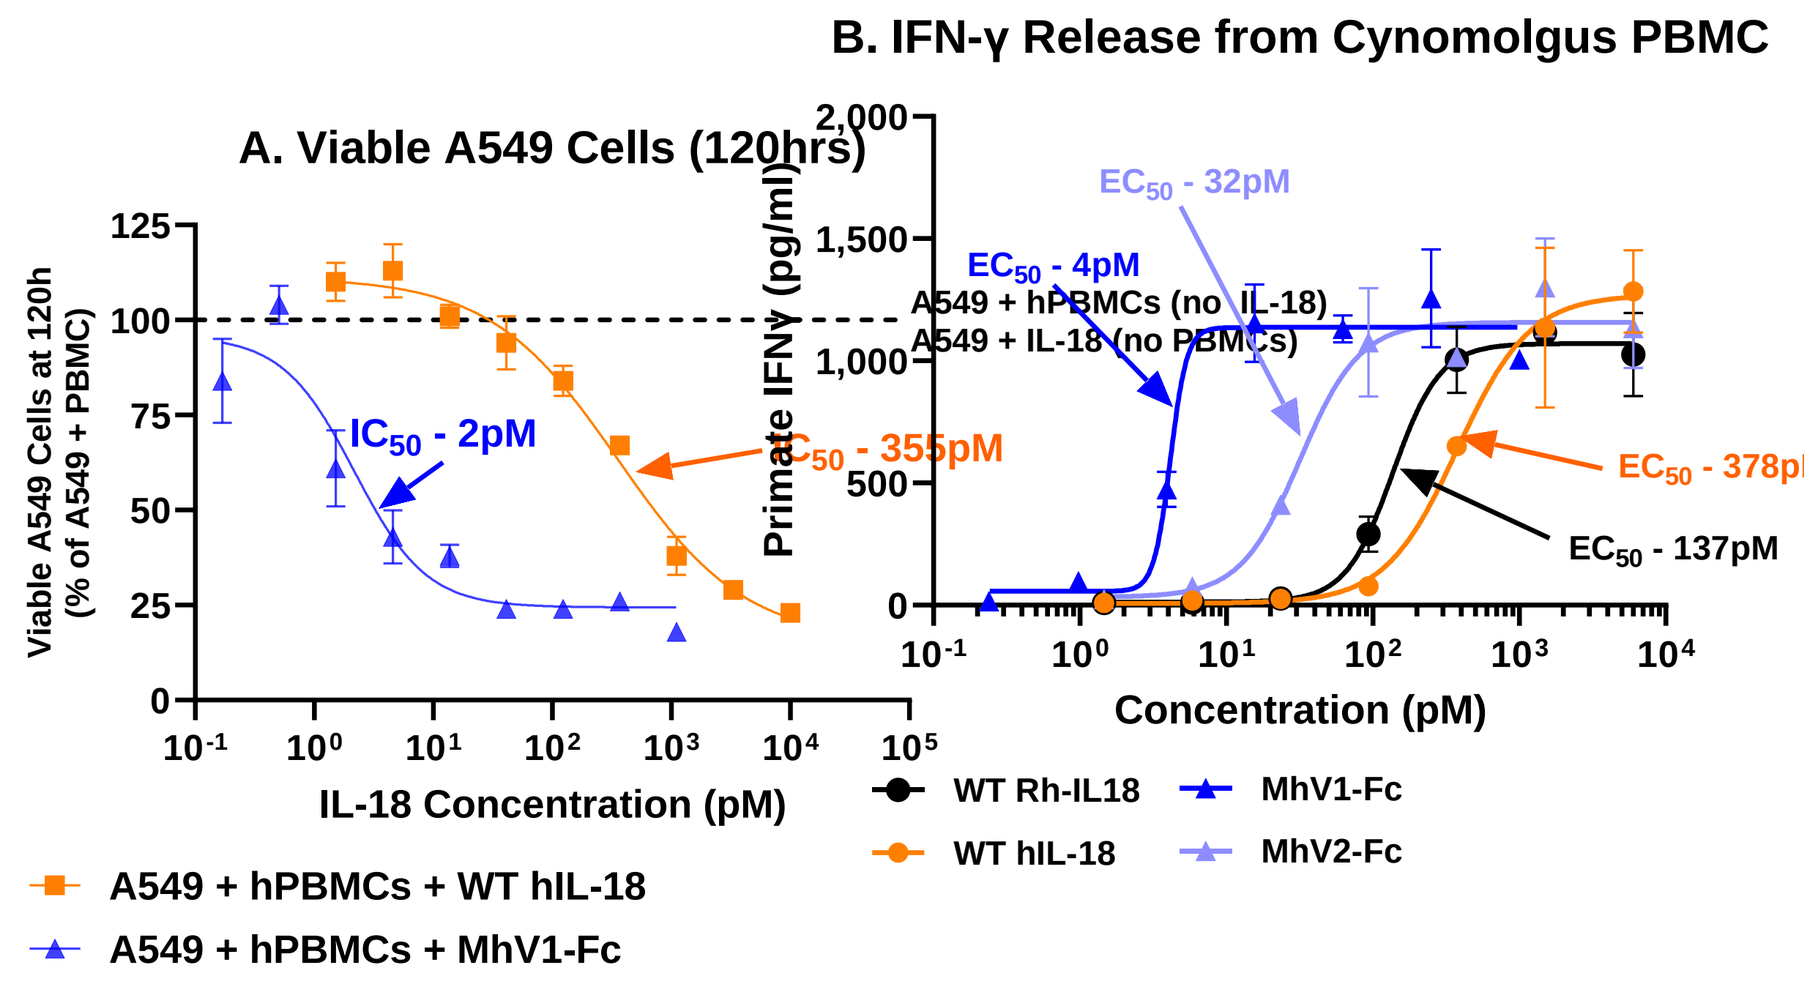
A. Viable A549 Cells (120hrs)
Viable A549 Cells at 120h (% of A549 + PBMC)
IC50 - 2pM
EC50 - 355pM
IL-18 Concentration (pM)
A549 + hPBMCs + WT hIL-18
A549 + hPBMCs + MhV1-Fc
B. IFN-γ Release from Cynomolgus PBMC
Primate IFNγ (pg/ml)
EC50 - 32pM
EC50 - 4pM
A549 + hPBMCs (no IL-18)
A549 + IL-18 (no PBMCs)
EC50 - 378pM
EC50 - 137pM
Concentration (pM)
WT Rh-IL18
WT hIL-18
MhV1-Fc
MhV2-Fc

A Half-life Enhanced Mouse Ortholog of Mural Human Variant-Fc Fusion Shows Durable Expansion and Activation of Cytotoxic NK Cells (Figure 2) and Effector CD8⁺ T Cells (Figure 3)

- A mouse ortholog to Mural hV-Fc fusion protein (mO-Fc) stimulated more durable expansion of blood NK and effector CD8⁺ T cells compared to mTV^{res}
- Upregulation of cytotoxic potential (Granzyme B⁺) of NK and effector CD8⁺ T cells was more durable for mO-Fc in comparison to mTV^{res}

Figure 2. *Expansion of Blood NK cells (NK1.1⁺) Following Subcutaneous Injection of Non-tumor Bearing C57BL/6 Mice with mTV^{res} or mO-Fc.* (A) Levels of blood NK1.1⁺ cells in C57BL/6 mice following subcutaneous injection of mTV^{res} or mO-Fc. (B) Cytotoxic phenotype of NK1.1⁺ cells in (A) as indicated by Granzyme B levels. The 0.64 mg/kg dose of mO-Fc is a close molar match to 0.32 mg/kg of mTV^{res}.

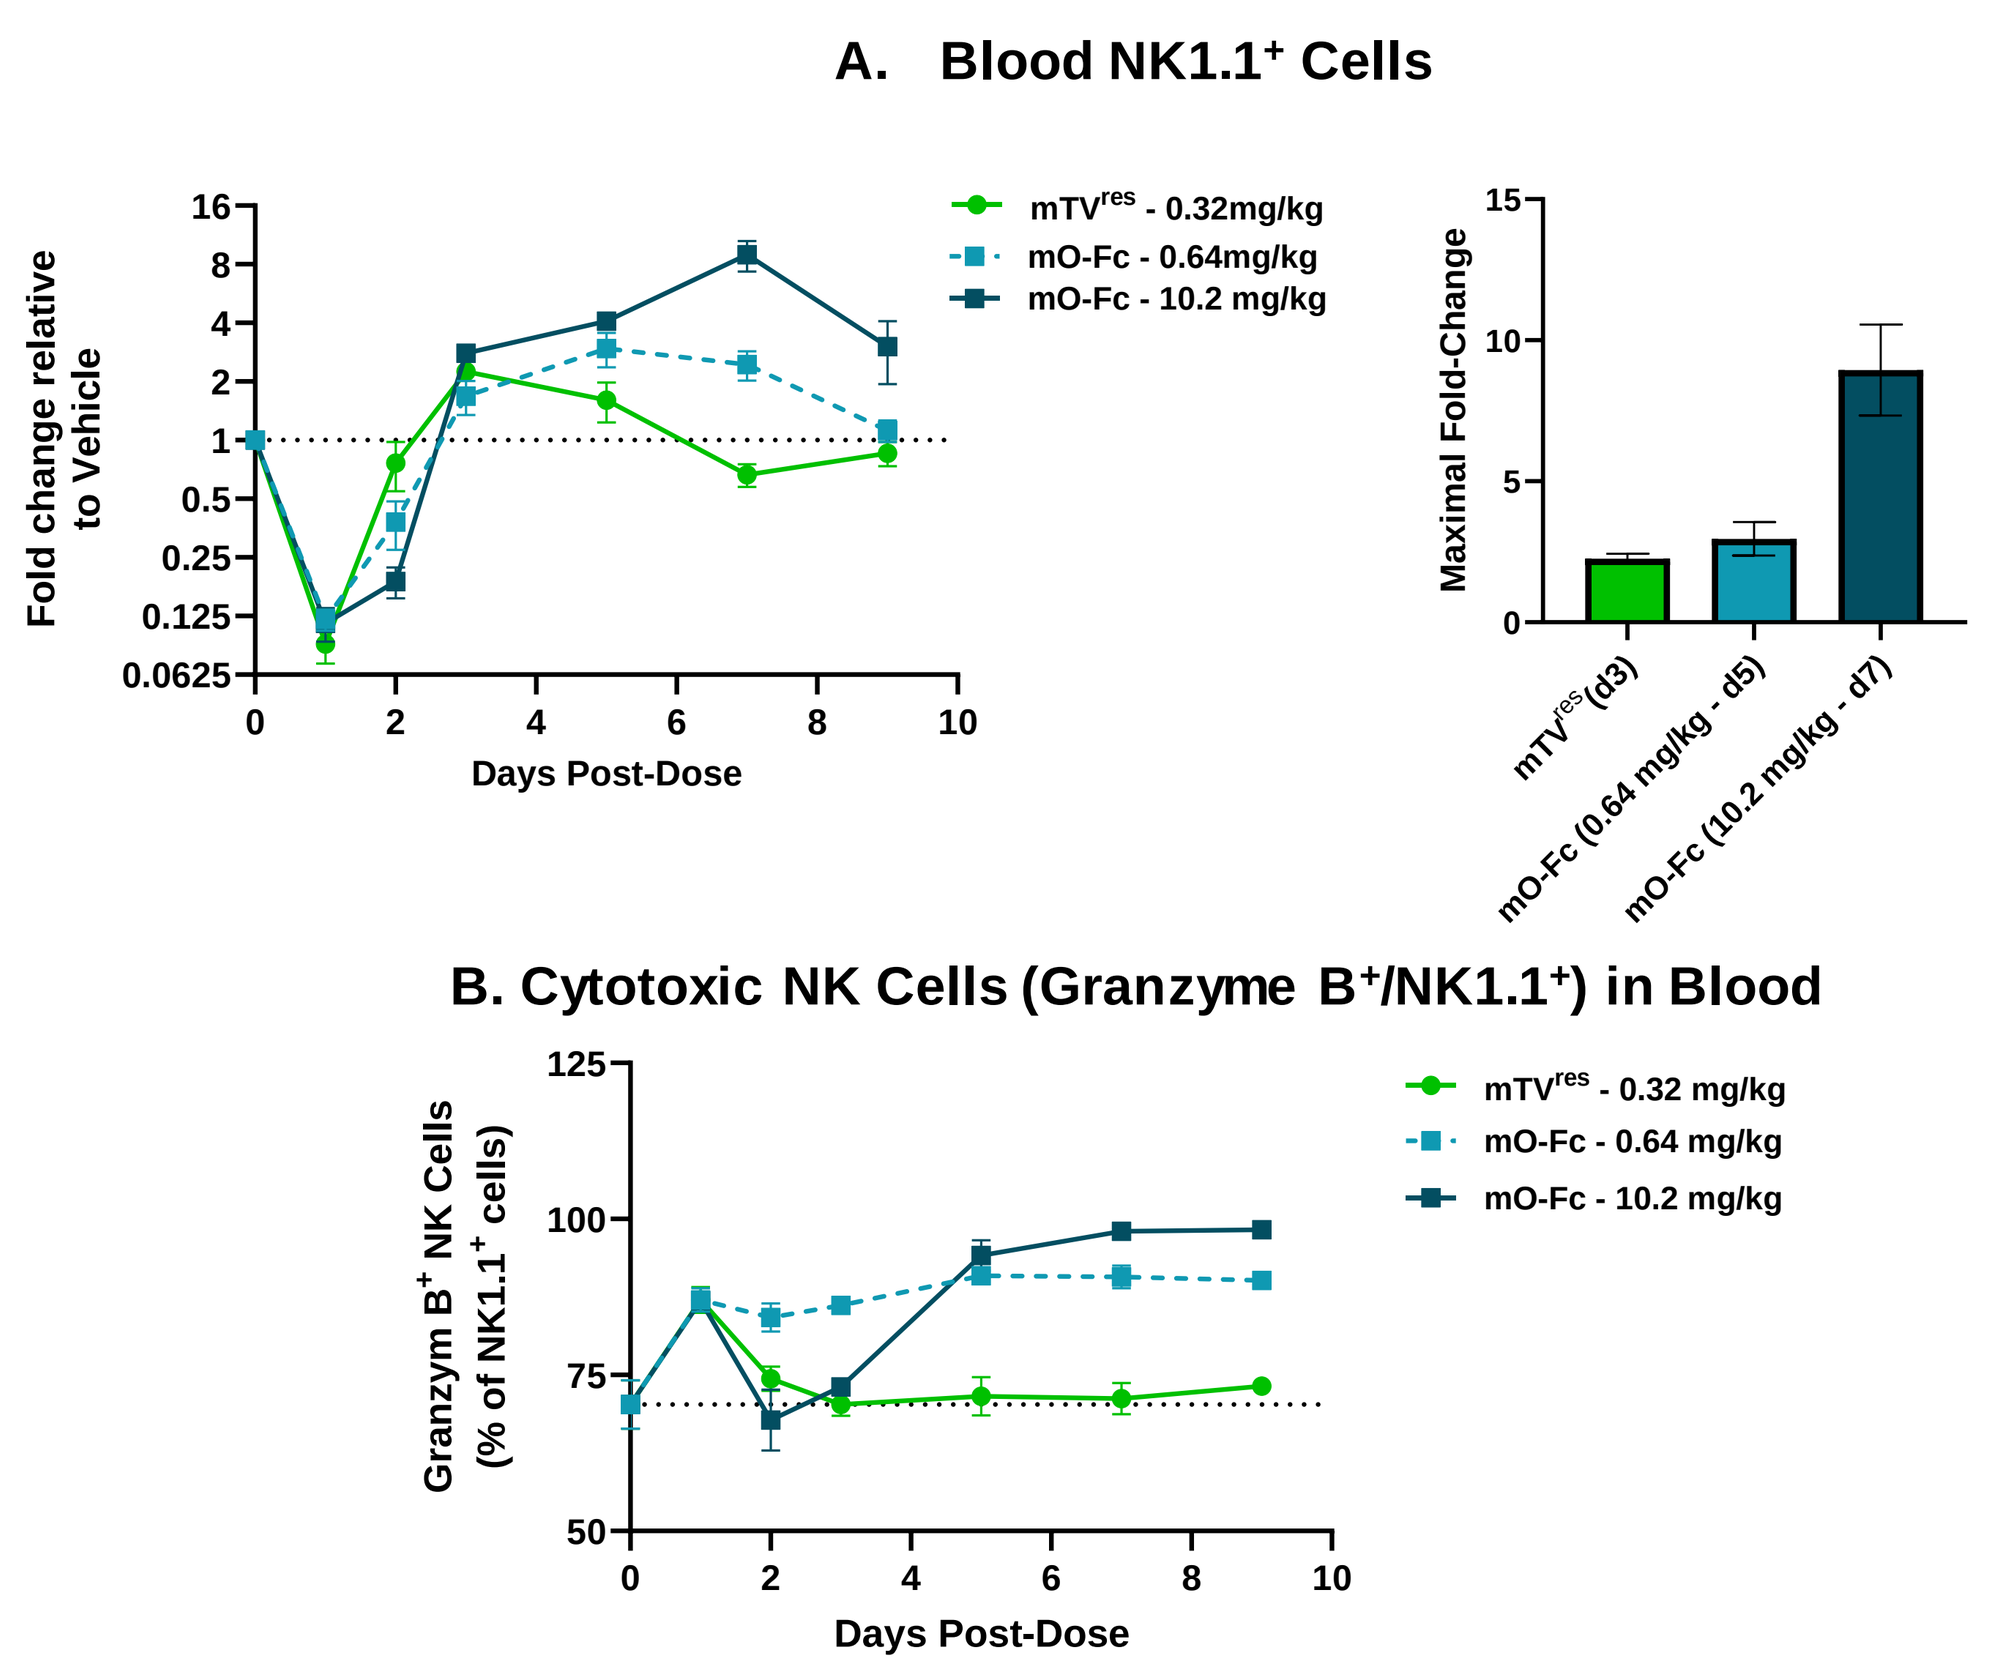
A. Blood NK1.1⁺ Cells
Fold change relative to Vehicle
Days Post-Dose
mTVres - 0.32mg/kg
mO-Fc - 0.64mg/kg
mO-Fc - 10.2 mg/kg
Maximal Fold-Change
mTVres (d3)
mO-Fc (0.64 mg/kg - d5)
mO-Fc (10.2 mg/kg - d7)
B. Cytotoxic NK Cells (Granzyme B⁺/NK1.1⁺) in Blood
Granzym B⁺ NK Cells (% of NK1.1⁺ cells)
Days Post-Dose
mTVres - 0.32 mg/kg
mO-Fc - 0.64 mg/kg
mO-Fc - 10.2 mg/kg

Figure 3. *Expansion of Blood Effector CD8⁺ T cells (CD62L⁻/CD44⁺) Following Subcutaneous Injection of mO-Fc in Non-tumor Bearing C57BL/6 Mice*. (A) Effector CD8⁺ T cells in blood of C57BL/6 mice treated as in Figure 2. (B) Activated phenotype of effector CD8⁺ T cells in (A) as indicated by Granzyme B and CD27 expression.

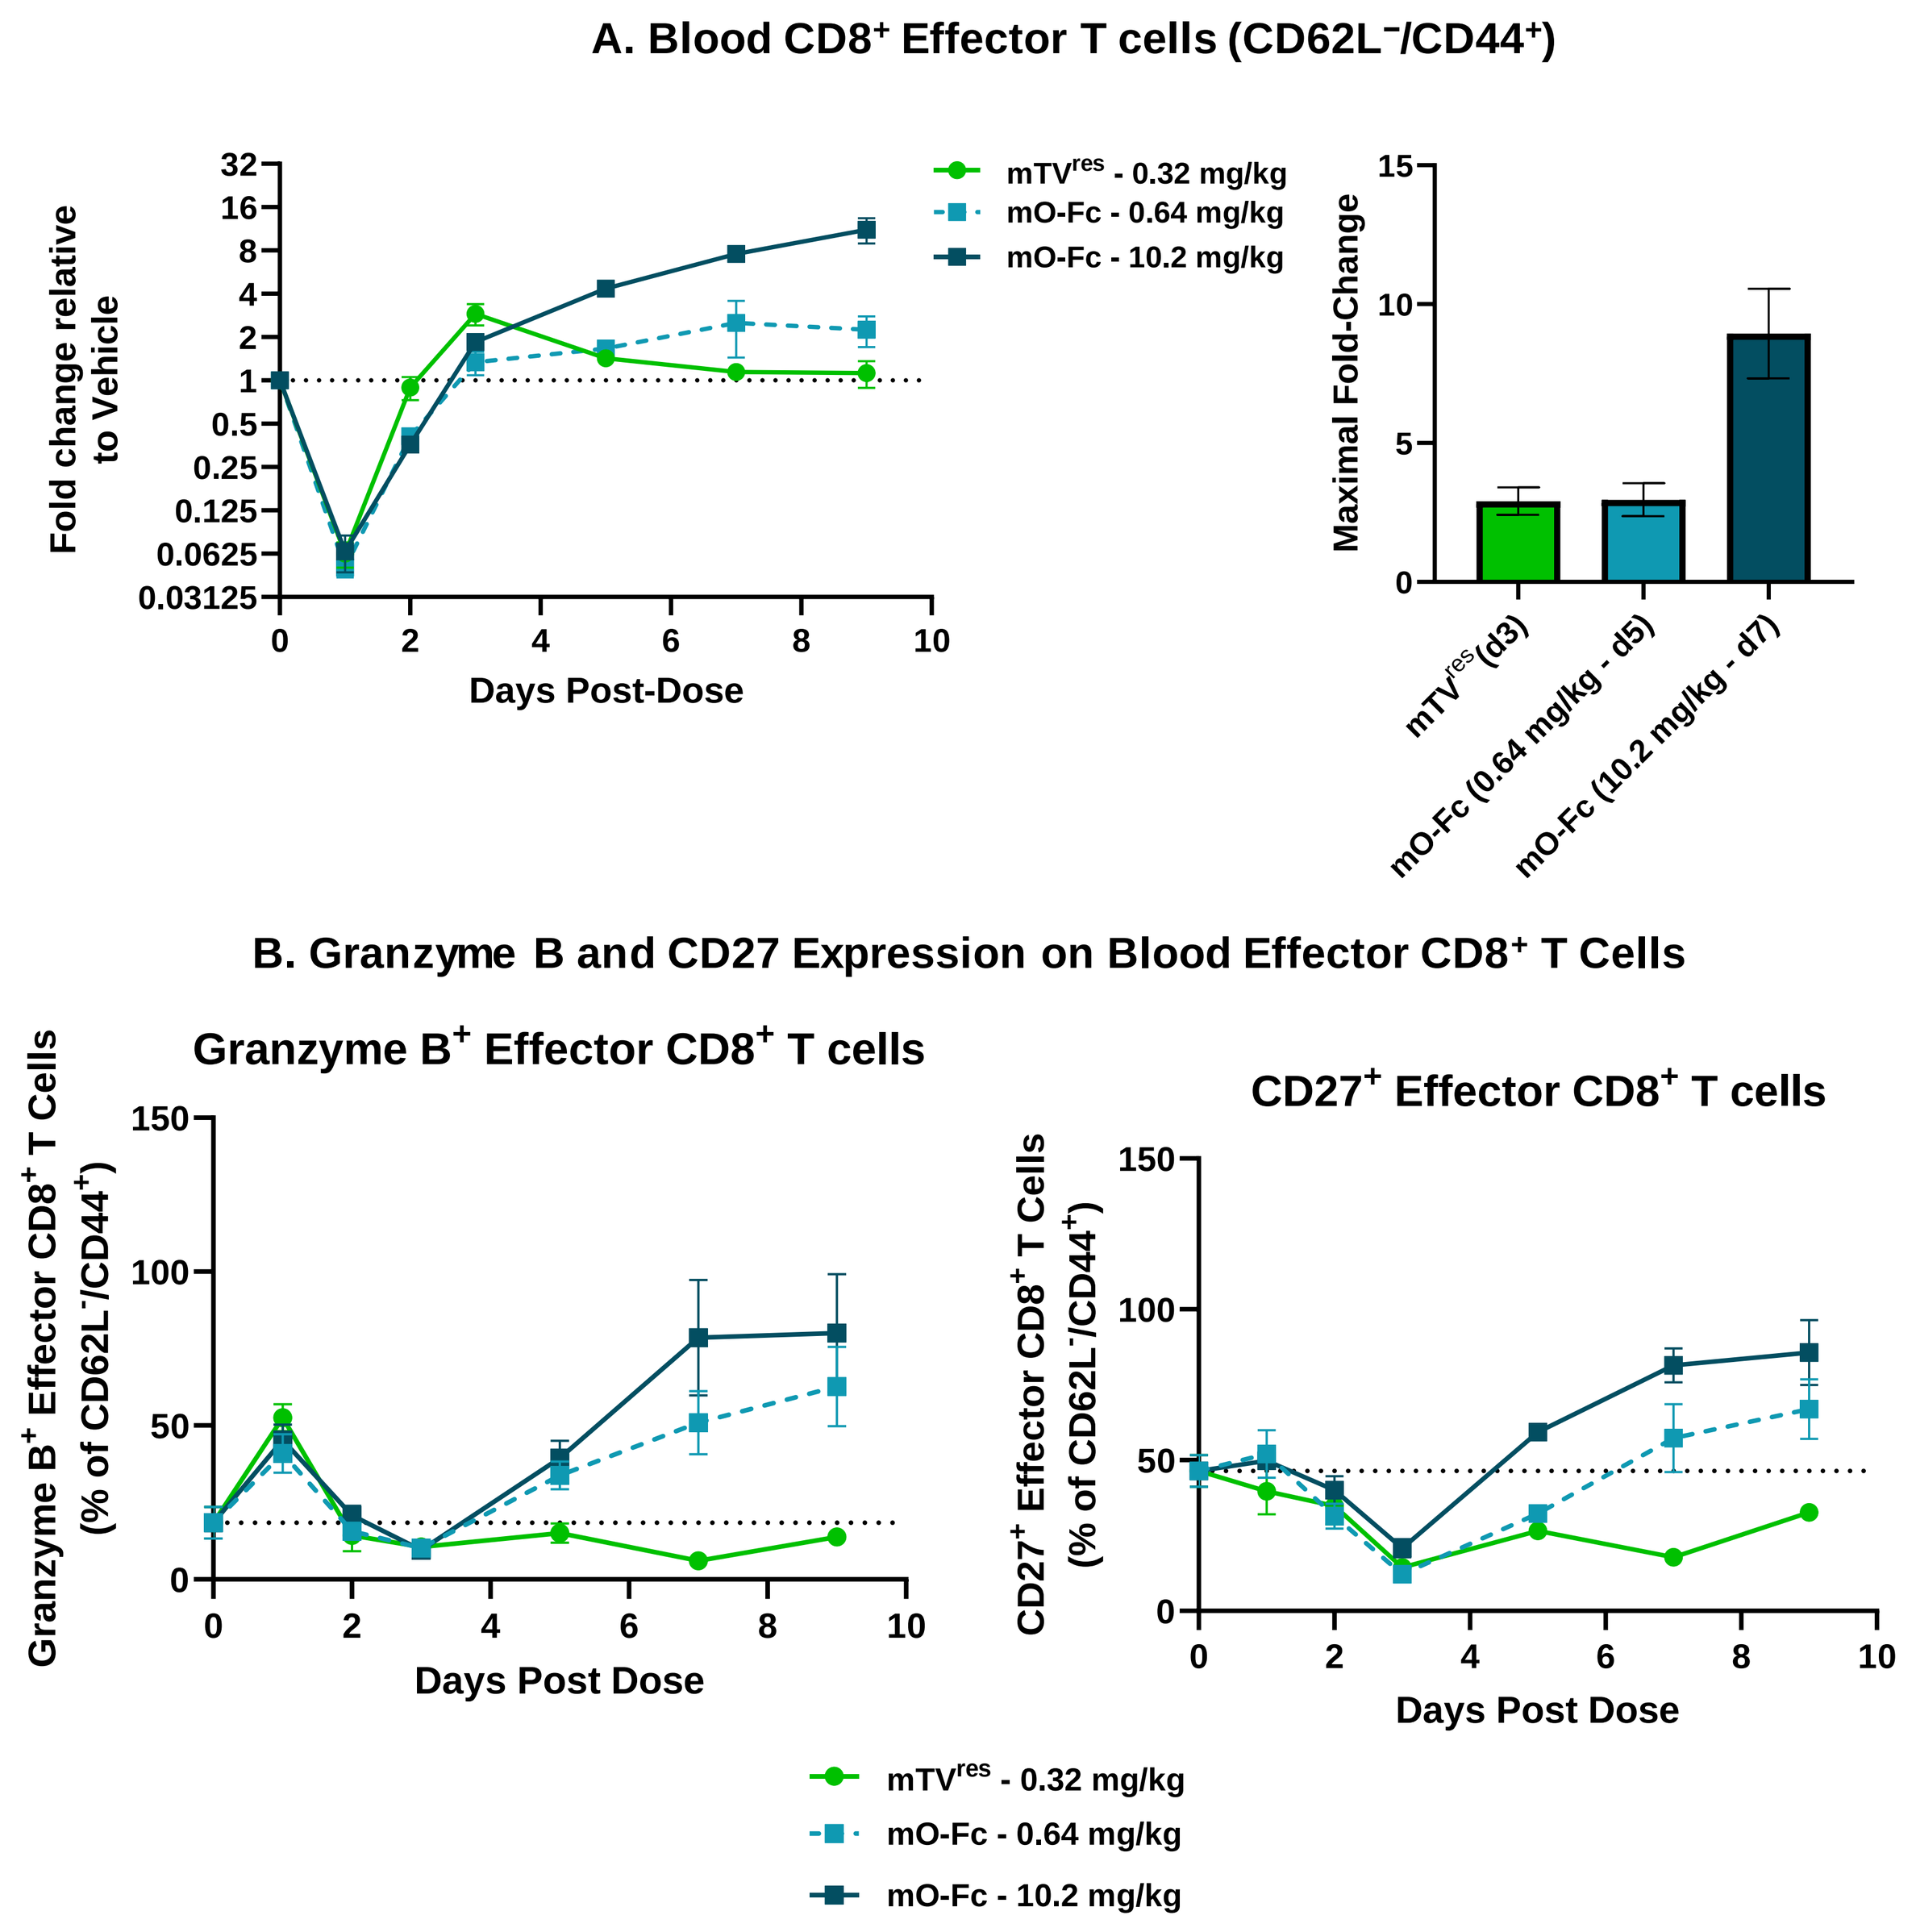
A. Blood CD8⁺ Effector T cells (CD62L⁻/CD44⁺)
Fold change relative to Vehicle
Days Post-Dose
mTVres - 0.32 mg/kg
mO-Fc - 0.64 mg/kg
mO-Fc - 10.2 mg/kg
Maximal Fold-Change
mTVres (d3)
mO-Fc (0.64 mg/kg - d5)
mO-Fc (10.2 mg/kg - d7)
B. Granzyme B and CD27 Expression on Blood Effector CD8⁺ T Cells
Granzyme B⁺ Effector CD8⁺ T cells
Granzyme B⁺ Effector CD8⁺ T Cells (% of CD62L⁻/CD44⁺)
Days Post Dose
CD27⁺ Effector CD8⁺ T cells
CD27⁺ Effector CD8⁺ T Cells (% of CD62L⁻/CD44⁺)
Days Post Dose
mTVres - 0.32 mg/kg
mO-Fc - 0.64 mg/kg
mO-Fc - 10.2 mg/kg

mO-Fc has Monotherapy Efficacy and Acceptable Safety Margin with q7d Dosing in the Heterotopic MC38 Syngeneic Model of Colorectal Cancer.

- mO-Fc demonstrated strong efficacy with dose-dependent increases in TGI, survival and complete responses
- A promising safety margin was observed with q7d dosing of mO-Fc
 - Strong efficacy and acceptable tolerability was observed over a broad range doses (0.64 mg/kg to 7.7mg/kg)

Figure 4. *Efficacy and Body Weight Tolerability of mO-Fc in Heterotopic MC38 Syngeneic Tumor Model.* Once weekly (q7d) dosing regimen (arrows) of the indicated dose levels of mO-Fc. Tumor growth inhibition (TGI, left panel), survival with complete responses (middle panel), and body weight (right panel). Plus sign (+) indicates subjects removed from study for moribund conditions unrelated to reaching the upper tumor volume limit. N=12 mice/group.

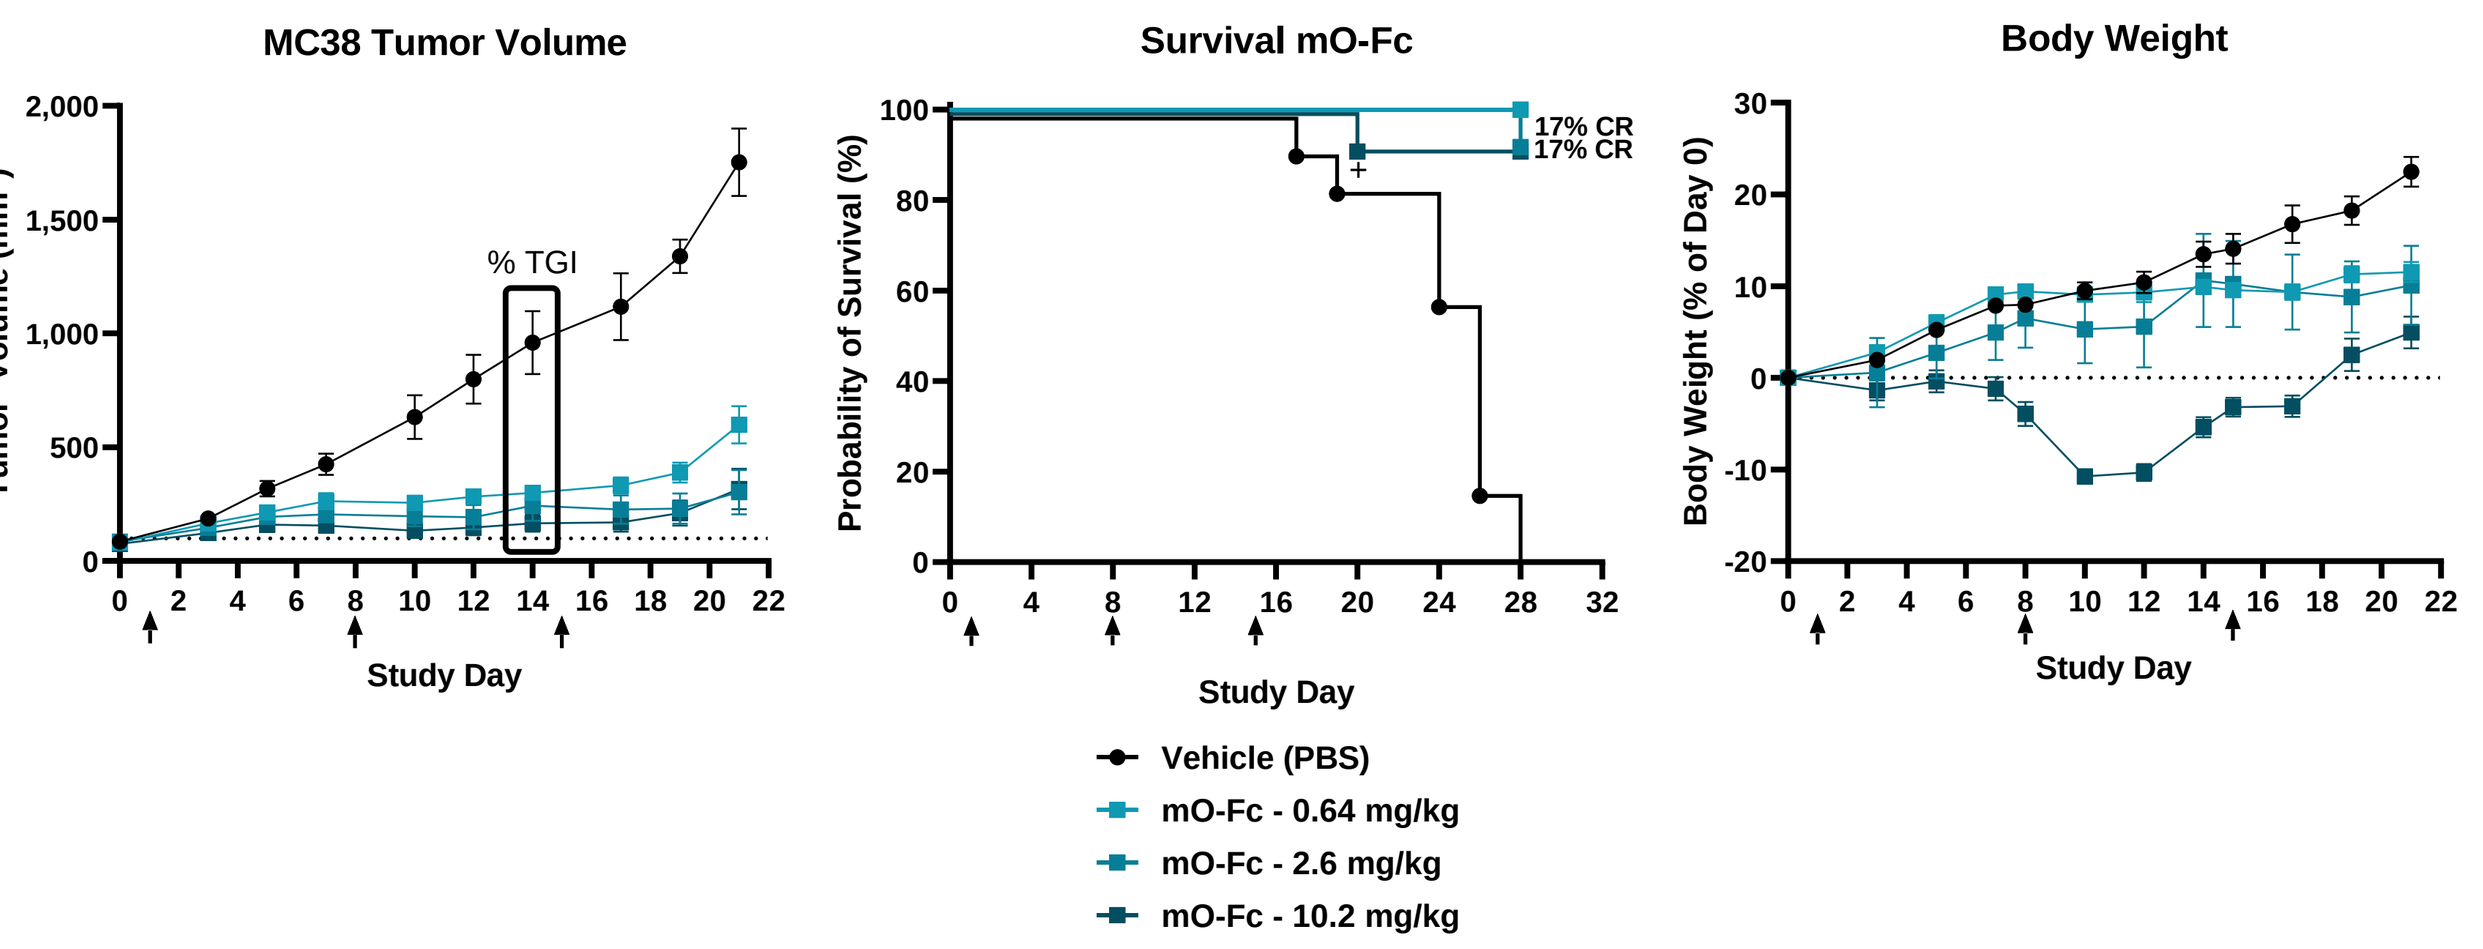
MC38 Tumor Volume
Tumor Volume (mm³)
% TGI
Study Day
Survival mO-Fc
Probability of Survival (%)
17% CR
17% CR
Study Day
Body Weight
Body Weight (% of Day 0)
Study Day
Vehicle (PBS)
mO-Fc - 0.64 mg/kg
mO-Fc - 2.6 mg/kg
mO-Fc - 10.2 mg/kg

Table 2. *Efficacy and Tolerability Observations at Indicated Dose Levels of mO-Fc in Heterotopic MC38 Syngeneic Tumor Model (Figure 4).*

Treatment (q7d)	Dose (mg/kg)	Tumor Growth Inhibition Day 14 (% Inhibition vs. Vehicle Group)	% Survival Day 28 (% Survival by Group)	% Complete Responses Day 28 (% of group)	Non-tumor Volume Related Mortality or Moribund Condition (% of group)
Vehicle (PBS)	-	N/A	0	0	0
mO-Fc	0.64	75	100	0	0
	2.6	82	92	17	0
	10.2	90	92	17	8

Figure 5. *Efficacy and Body Weight Tolerability of mO-Fc and mTV^{sens}-Fc in MC38 Syngeneic Tumor Model.* A second efficacy study performed as described in Figure 3 with dosing groups of mO-Fc (blue shaded squares) and an IL-18BP-sensitive, half-life enhanced mouse tool variant mTV^{sens}-Fc (red circles). N=10 mice/group.

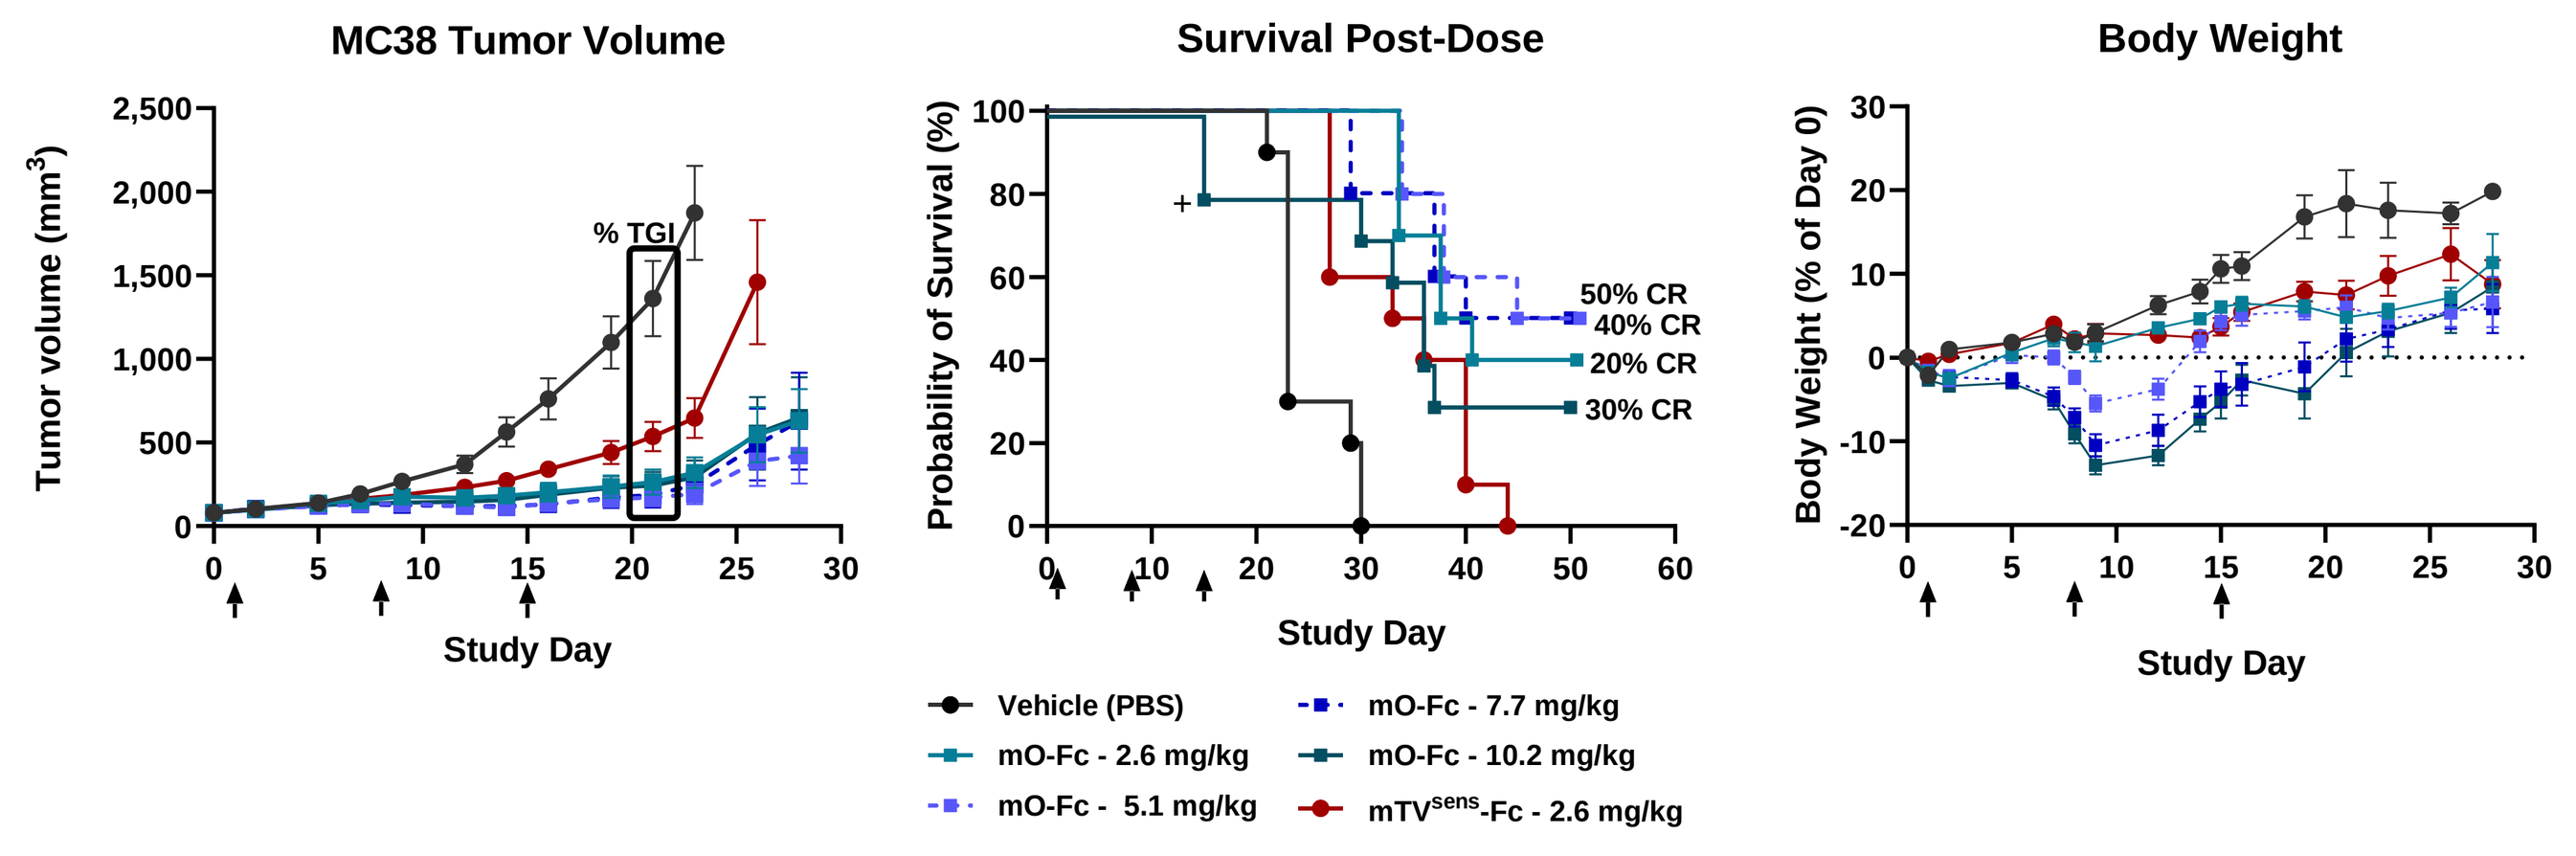
MC38 Tumor Volume
Tumor volume (mm³)
% TGI
Study Day
Survival Post-Dose
Probability of Survival (%)
50% CR
40% CR
20% CR
30% CR
Study Day
Body Weight
Body Weight (% of Day 0)
Study Day
Vehicle (PBS)
mO-Fc - 2.6 mg/kg
mO-Fc - 5.1 mg/kg
mO-Fc - 7.7 mg/kg
mO-Fc - 10.2 mg/kg
mTVsens-Fc - 2.6 mg/kg

Table 3. *Efficacy and Tolerability Observations for mO-Fc and mTV^{sens}-Fc in Heterotopic MC38 Syngeneic Tumor Model (Figure 5).*

Treatment (q7d)	Dose (mg/Kg)	Tumor Growth Inhibition Day 21 (% Inhibition vs. Vehicle Group)	% Survival Day 50 (% Survival by group)	% Complete Responses Day 50 (% of group)	Non-tumor Volume Related Mortality or Moribund Condition (% of group)
Vehicle (PBS)	-	N/A	0	0	0
mO-Fc	2.6	86	40	20	0
	5.1	93	50	40	0
	7.7	92	50	50	0
	10.2	87	30	30	20
mTV^{sens}-Fc	2.6	64	0	0	0

References

1. Whitmore et al (2024). *Interleukin-18 engineered for resistance to IL-18 binding protein (IL-18BP) and half-life extension to enhance its therapeutic potential.* AACR Annual Meeting 2024. Abstract # 4076.

Acknowledgments

- Vicki Goodman, Eric Guan, Maiken Keson-Brookes and Matthew Salkovitz for editorial review

SITC 39th Annual Meeting, Houston, TX • November 6-10, 2024